Single and multiple dose safety, tolerability and pharmacokinetics of the selective M_1 receptor partial agonist HTL0018318 in healthy volunteers

Tim Tasker MBBS[1], Jan Liptrot PhD[1], Charlotte Bakker MD[2], Ellen 't Hart PhD[2], Erica Klaassen PhD[2], Samantha Prins MD[2], Thalia van der Doef PhD[2], Mike Walker[2], Giles A. Brown PhD[1], Alastair Brown PhD[1], Miles Congreve PhD[1], Malcolm Weir PhD[1], Fiona H. Marshall PhD[1], David M. Cross PhD[4], Geert Jan Groeneveld MD, PhD[2], Pradeep. J. Nathan PhD[1,3]

[1]Sosei Heptares, Cambridge UK; [2]Centre for Human Drug Research (CDHR), Leiden, Netherlands; [3]Department of Psychiatry, University of Cambridge, UK; [4]Cross Pharma Consulting Limited, Cambridge, UK

Background

- The cholinergic neurons of the basal forebrain and medial septum provide the major source of cholinergic innervation to the neocortex and hippocampus and play a critical role modulating cognitive processes such as learning and memory, in part through activation of post-synaptic M_1 receptors.
- It is widely accepted that Alzheimer's disease (AD) is associated with significant early and progressive loss of cholinergic neurons. Cholinesterase inhibitors including donepezil have modest efficacy, potentially because they target degenerating pre-synaptic cholinergic neurons.
- An alternative and potentially more effective strategy is to target post-synaptic M_1 receptors which are relatively preserved in AD (Figure 1).
- Muscarinic receptor agonists including the M_1/M_4 agonist xanomeline and the M_1 orthosteric agonist GSK1034702 have shown promising early clinical effects but were not further developed due to gastrointestinal and cardiovascular adverse events (AEs).
- HTL0018318 is a selective M_1 agonist that was developed using the Heptares StaR® technology – an integrated structure/ chemistry/ pharmacology platform (SBDD) (Figure 2).
- HTL0018318 is currently under development for the symptomatic treatment of cognitive impairment in dementias including AD and Dementia with Lewy bodies (DLB).
- The primary objective: To examine the single and multiple dose safety, tolerability, pharmacokinetics (PK) of HTL0018318 in healthy younger adult and elderly subjects. Pharmacodynamic biomarkers were also assessed but are not presented here.

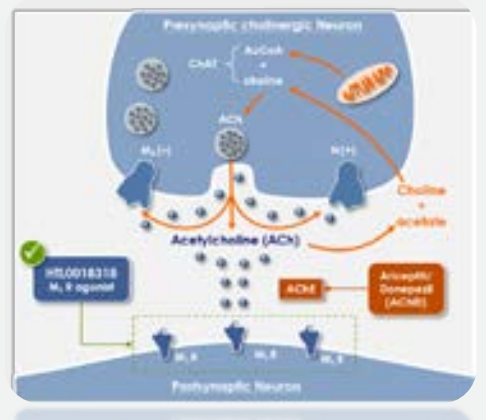
Presynaptic cholinergic Neuron
Acetylcholine (ACh)
HTL0018318 M_1 R agonist
AChE
Postsynaptic Neuron

Figure 1: HTL0018318 binds to post-synaptic M_1 receptors on cholinergic neurons in the cortex and hippocampus

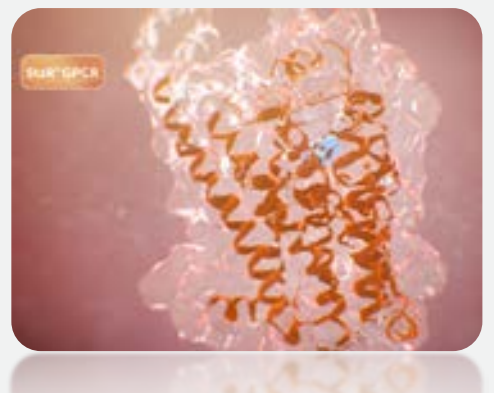

Figure 2: StaR® technology. Single point mutations stabilize GPCRs to create a StaR® that can be purified and retains function

Methods

- The single ascending dose (SAD) study was a single centre, randomized, double-blind, placebo-controlled, sequential, single ascending oral (solution) dose study.

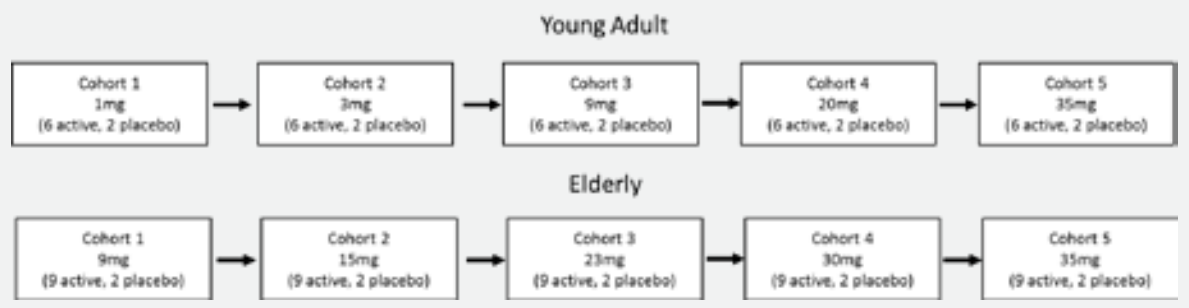
Young Adult
Cohort 1 1mg (6 active, 2 placebo)
Cohort 2 3mg (6 active, 2 placebo)
Cohort 3 9mg (6 active, 2 placebo)
Cohort 4 20mg (6 active, 2 placebo)
Cohort 5 35mg (6 active, 2 placebo)
Elderly
Cohort 1 9mg (9 active, 2 placebo)
Cohort 2 15mg (9 active, 2 placebo)
Cohort 3 23mg (9 active, 2 placebo)
Cohort 4 30mg (9 active, 2 placebo)
Cohort 5 35mg (9 active, 2 placebo)

- The multiple ascending dose (MAD) study was a single centre, randomised, double-blind, placebo-controlled, sequential, multiple ascending oral (solution) dose study.

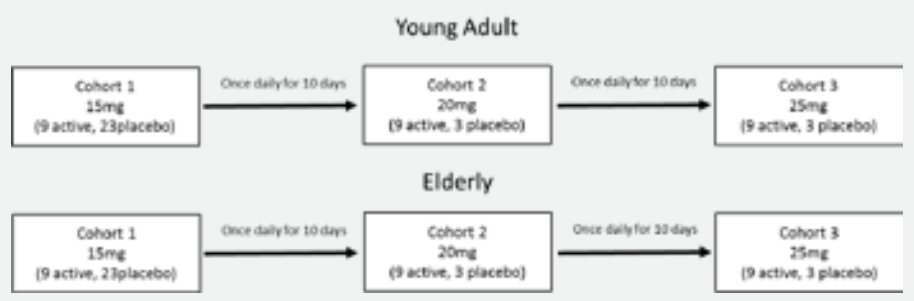
Young Adult
Cohort 1 15mg (9 active, 23placebo)
Once daily for 10 days
Cohort 2 20mg (9 active, 3 placebo)
Once daily for 10 days
Cohort 3 25mg (9 active, 3 placebo)
Elderly
Cohort 1 15mg (9 active, 23placebo)
Once daily for 10 days
Cohort 2 20mg (9 active, 3 placebo)
Once daily for 10 days
Cohort 3 25mg (9 active, 3 placebo)

- One elderly cohort was initially dosed at 35mg/day (2 active) but due to lack of tolerability, subsequently dosed at 35mg using a titration regimen (5 days on 20mg/day and 10 days on 35mg/day) (7 active, 3 placebo).

Results

Single Doses (SAD study)

- PK of HTL0018318 were well-characterized in all subjects after single doses (Figure 3).
 - Rapid absorption was rapid with a typical Tmax of 1.0-1.5h post-dose.
 - Apparent mean half-life of 12-16h.
 - Distribution into CSF (CSF:plasma ratio≈30%).
 - No food effect on AUC or half-life
- Single doses of HTL0018318 were associated with mild dose-related AEs (with low incidence) in both younger and elderly subjects.
 - Most frequently reported cholinergic related AEs included hypersalivation, hyperhidrosis and increases in blood pressure, particularly following the 35mg dose (younger adult) and 23mg and 35mg doses (elderly).
- In younger adult subjects, doses up to 20mg (compared to placebo) were not associated with changes in systolic and diastolic blood pressure and heart rate.
 - 35mg dose (compared to placebo) was associated with an increase in mean systolic and diastolic blood pressure (up to 10mmHg) and mean heart rate (up to 9.8bpm).
- In elderly subjects, significant increases in mean systolic and diastolic blood pressure (up to 11.9mmHg) and mean heart rate (up to 6.3bpm) were observed in the 15-35mg dose range.
 - No clear evidence of dose-dependency.

Multiple Doses (MAD study)

- PK of HTL0018318 were well-characterized in all subjects after multiple doses.
 - Consistent with single dose PK and showing no meaningful change over time.
 - Moderate inter-individual variability in exposure (6-46 %CV for Cmax and AUC0-24h).
- HTL0018318 in repeated administration up to 35 mg/day for 10 days was generally well-tolerated, with mild AEs (with low incidence) and some evidence for dose-dependency. The most frequently reported cholinergic related AEs are shown in Table 1 and 2.
- Repeated administration HTL0018318 over 10 days was associated with some small statistically significant increases in blood pressure on day 1 (up to 8.7mmHg) compared to placebo with a decline in this difference with continued dosing (Figure 4).
 - No consistent or clear dose-response relationships.
- HTL0018318 caused small increases in mean heart rate (up to 10bpm) (Figure 5).
 - increases were in the context of overall decreases in mean heart rate (i.e. smaller decreases with HTL0018318 relative to the placebo decrease).
- There were no clinically significant changes in blood and urine laboratory values or abnormalities in the ECGs and Holter assessments following both single and multiple ascending doses up to 35mg.

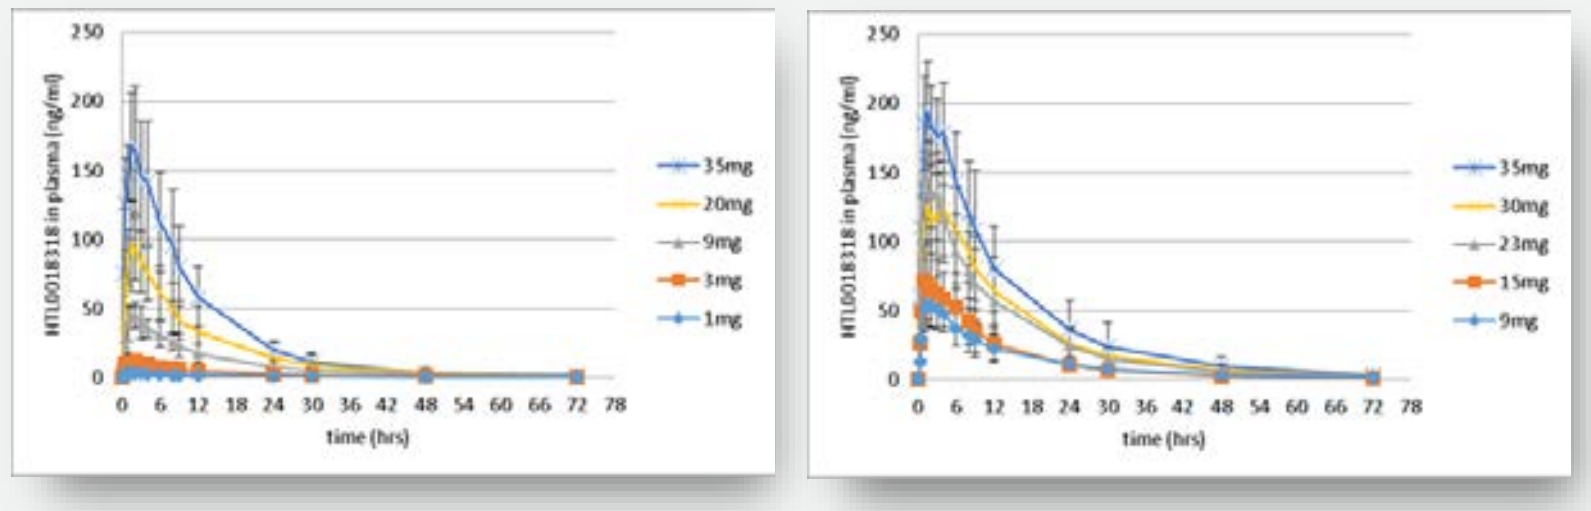
HTL0018318 in plasma (ng/ml)
time (hrs)
35mg
20mg
9mg
3mg
1mg
35mg
30mg
23mg
15mg
9mg

Figure 3. HTL0018318 arithmetic mean (± standard deviation) plasma concentration against time after dose following single oral doses of HTL0018318 in healthy young (Left) and elderly (Right) subjects.

TEAE	Adults 15 mg (n=9)	Adults 20 mg (n=9)	Adults 25 mg (n=9)	Adults placebo (n=9)
Hypertension	0	0	0	0
Nausea	3	2	4	0
Vomiting	0	0	1	0
Diarrhoea	1	1	0	0
Constipation	0	0	0	0
Hypersalivation	0	0	1	0
Insomnia	0	0	0	1
Dizziness	0	1	1	0
Muscle spasm	1	0	0	0
Hyperhidrosis	1	4	3	0
Cold sweat	0	1	0	0
piloerection	0	2	0	0
Chills	1	0	0	0
Feeling cold	0	1	0	0
Feeling hot	0	1	0	0
Feeling of body temperature change	0	0	0	0
Hot flush	0	1	2	0
Peripheral coldness	0	0	0	0
Total number of subjects	5	5	6	1

Table 1. Number of subjects that reported possible cholinergic AEs – Young adults

TEAE	Elderly 15 mg (n=9)	Elderly 20 mg (n=9)	Elderly 25 mg (n=9)	Elderly 35 mg (n=2)	Elderly 35 mg/up-titration (n=7)	Elderly placebo (n=14)
Hypertension	0	0	1	1	1	0
Nausea	1	1	2	0	0	0
Vomiting	0	0	0	0	0	0
Diarrhoea	0	0	1	0	0	0
Constipation	0	0	0	0	0	1
Hypersalivation	0	0	0	0	0	0
Insomnia	0	1	0	0	0	0
Dizziness	0	0	3	0	0	1
Muscle spasm	0	1	0	0	0	0
Hyperhidrosis	0	3	5	0	2	0
Cold sweat	0	0	0	1	2	0
piloerection	0	0	0	0	0	0
Chills	0	0	1	0	4	0
Feeling cold	0	1	2	1	2	1
Feeling hot	0	0	2	0	0	0
Feeling of body temperature change	0	0	1	0	0	0
Hot flush	0	2	1	0	2	0
Peripheral coldness	0	0	0	1	0	0
Total number of subjects	1	5	7	2	7	2

Table 2. Number of subjects that reported possible cholinergic AEs – Elderly subjects

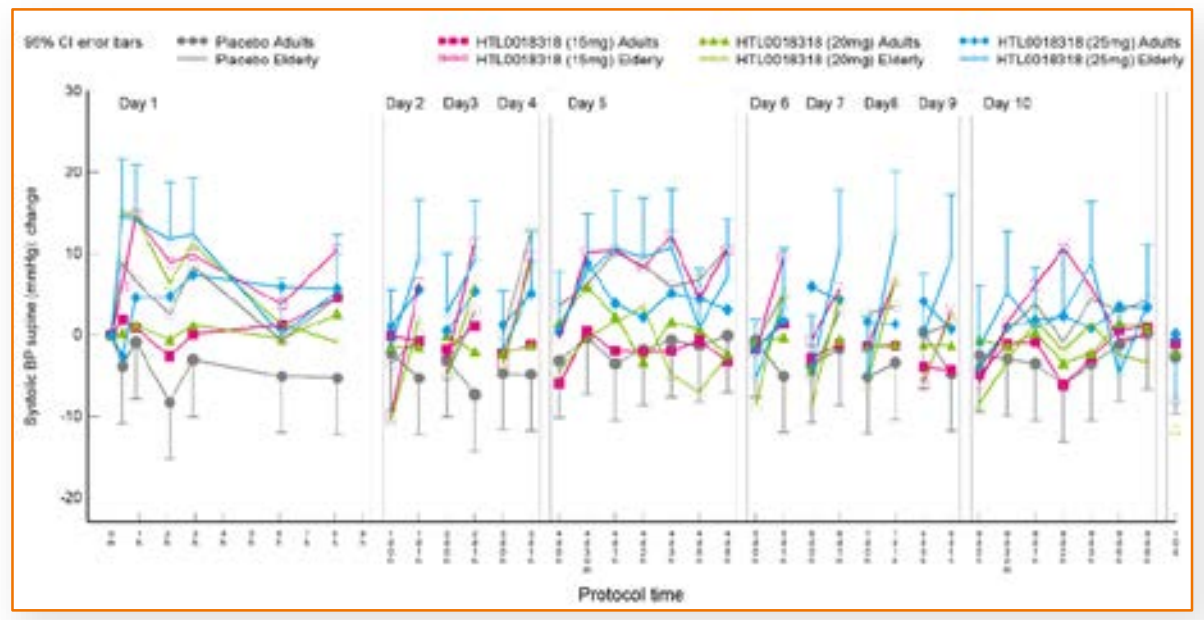
95% CI error bars
Placebo Adults
Placebo Elderly
HTL0018318 (15mg) Adults
HTL0018318 (15mg) Elderly
HTL0018318 (20mg) Adults
HTL0018318 (20mg) Elderly
HTL0018318 (25mg) Adults
HTL0018318 (25mg) Elderly
Day 1
Day 2
Day3
Day 4
Day 5
Day 6
Day 7
Day8
Day 9
Day 10
Systolic BP supine (mmHg): change
Protocol time

Figure 4. LSM Change from baseline systolic blood pressure supine (mm Hg) over 10 days dosing

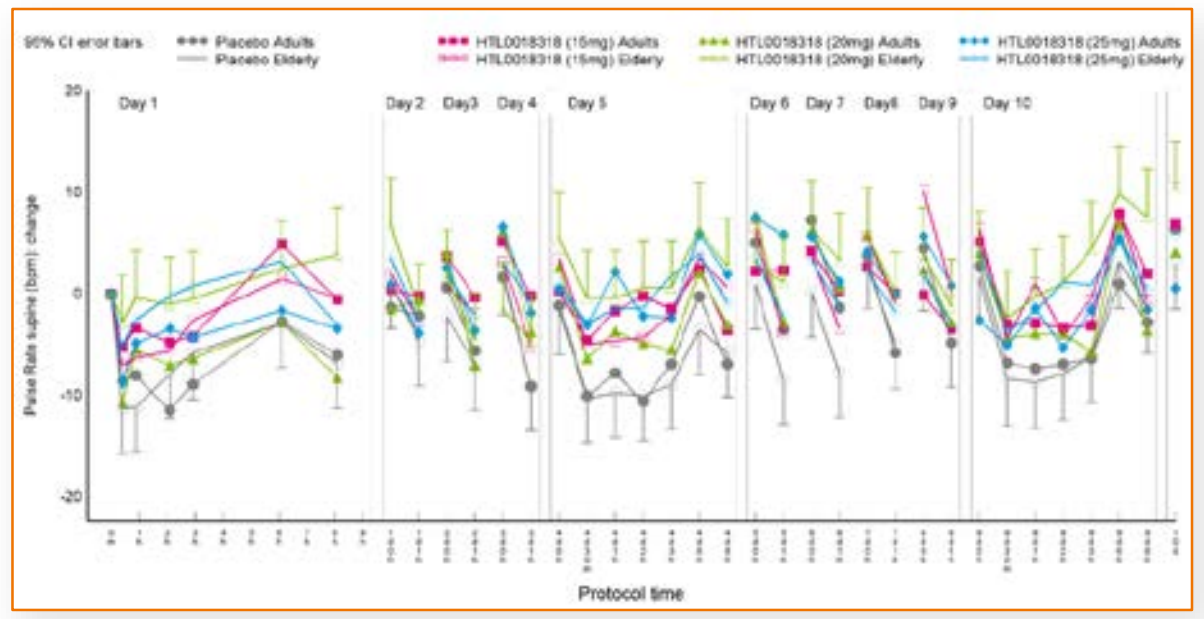
95% CI error bars
Placebo Adults
Placebo Elderly
HTL0018318 (15mg) Adults
HTL0018318 (15mg) Elderly
HTL0018318 (20mg) Adults
HTL0018318 (20mg) Elderly
HTL0018318 (25mg) Adults
HTL0018318 (25mg) Elderly
Day 1
Day 2
Day3
Day 4
Day 5
Day 6
Day 7
Day8
Day 9
Day 10
Pulse Rate supine (bpm): change
Protocol time

Figure 5. LSM Change from baseline pulse rate supine (bpm) over 10 days

Conclusions

- HTL0018318 showed well characterised pharmacokinetics in young adults and elderly subjects following single and multiple doses over 10 days.
- HTL0018318 was generally well tolerated in the dose range studied.
- The initial increase in blood pressure following single doses tended to decline with repeated dosing while increases in heart rate were small relative to baseline.
- These findings provide encouraging safety and pharmacokinetic data in support of the development of HTL0018318 as a symptomatic treatment for cognitive impairment in dementias including AD and DLB.

sosei HEPTARES

Single and multiple dose safety, tolerability and pharmacokinetics of the selective M_1 receptor partial agonist HTL0018318 in healthy volunteers

Tim Tasker MBBS[1], Jan Liptrot PhD[1], Charlotte Bakker MD[2], Ellen 't Hart PhD[2], Erica Klaassen PhD[2], Samantha Prins MD[2], Thalia van der Doef PhD[2], Mike Walker[2], Giles A. Brown PhD[1], Alastair Brown PhD[1], Miles Congreve PhD[1], Malcolm Weir PhD[1], Fiona H. Marshall PhD[1], David M. Cross PhD[4], Geert Jan Groeneveld MD, PhD[2], Pradeep. J. Nathan PhD[1,3]

[1]Sosei Heptares, Cambridge UK; [2]Centre for Human Drug Research (CDHR), Leiden, Netherlands; [3]Department of Psychiatry, University of Cambridge, UK; [4]Cross Pharma Consulting Limited, Cambridge, UK

Safety Update on HTL0018318

- Sosei Heptares and Allergan, its license partner for HTL0018318, have decided to voluntarily suspend clinical development activities with HTL0018318 pending the investigation of an unexpected toxicology finding in an animal study involving non-human primates. This voluntary suspension is not based on any human findings.
- To date, HTL0018318 has been investigated in approximately 310 human subjects in the US and Europe, including healthy volunteers and patients with mild/moderate AD. Data available from the human studies have found it to be well tolerated and with no serious adverse effects at the tested doses for up to 28 days. Patient safety is of the utmost importance to Sosei Heptares and Allergan. The decision by Sosei Heptares and Allergan to voluntarily suspend clinical development activities with HTL0018318 was taken as a precaution until the completion of further investigation.

sosei
HEPTARES