Efficacy and safety of once-daily low-dose indacaterol/mometasone via Breezhaler® in symptomatic adult and adolescent patients with inadequately controlled asthma: Phase III randomised QUARTZ study findings

Peter D'Andrea[1], Oliver Kornmann[2], Janos Mucsi[3], Motokazu Kato[4], Lorraine Bandelli[1], Biswajit Sen[5]

[1]Novartis Pharmaceuticals Corporation, East Hanover, NJ, United States; [2]IKF Pneumologie Frankfurt, Clinical Research Centre Respiratory Diseases, Frankfurt, Germany; [3]Erzsébet Gondozóház, Gödöllő, Hungary; [4]Chest Disease Clinical and Research Institute, Kishiwada City Hospital, Kishiwada, Japan; [5]Novartis Healthcare Pvt. Ltd, Hyderabad, India

RCT3780

Introduction

- Global initiative for asthma (GINA) 2019 recommends the addition of long-acting β_2-agonist (LABA) to inhaled corticosteroid (ICS) monotherapy as the maintenance controller therapy in patients with inadequately controlled asthma[1]
- The combination helps in achieving asthma control by reducing symptoms and improving lung function in these patients[1]
- Indacaterol acetate/mometasone furoate (IND/MF, a LABA/ICS combination) 150/80 µg once-daily (o.d.) delivered via Breezhaler®, a single-dose dry powder inhaler, is a fixed-dose combination (FDC) of IND (LABA) and MF (a low-dose ICS), and is under development for treatment of symptomatic adult and adolescent patients with inadequately controlled asthma
- Here, we present the results from QUARTZ study
- The objective of the study was to evaluate efficacy and safety of IND/MF (150/80 µg o.d.) delivered via Breezhaler® compared with MF 200 µg o.d. delivered via Twisthaler® in terms of lung function (trough forced expiratory volume in one second [FEV_1]) and asthma control (asthma control questionnaire [ACQ-7] score) in inadequately controlled asthma patients

Methods

Patients

Key inclusion criteria

- Men and women (adult and adolescent patients) aged ≥12 and ≤75 years
- Diagnosis of asthma for a period of ≥3 months prior to screening
- Patients who were taking low-dose ICS (with or without LABA) prior to the study visit for ≥1 month
- ACQ-7 ≥1.5 prior to randomisation
- Pre-bronchodilator FEV_1 ≥60% and <90% of the predicted normal value according to ATS/ERS criteria after withholding bronchodilators at the start and end of run-in period

Key exclusion criteria

- Patients who had an asthma attack/exacerbation requiring systemic steroids or hospitalisation or emergency room visit within the past 6 weeks for adults (6 months for adolescents)
- Patients with history of a long QT syndrome, and a prolonged QTc interval
- History of chronic lung disease other than asthma

Study design

- The QUARTZ study was a Phase III, multicentre, randomised, double-blind, double-dummy and parallel-group study that included a 12-week treatment period (Clinical trials identifier: NCT02892344; **Figure 1**)

Figure1. QUARTZ study design

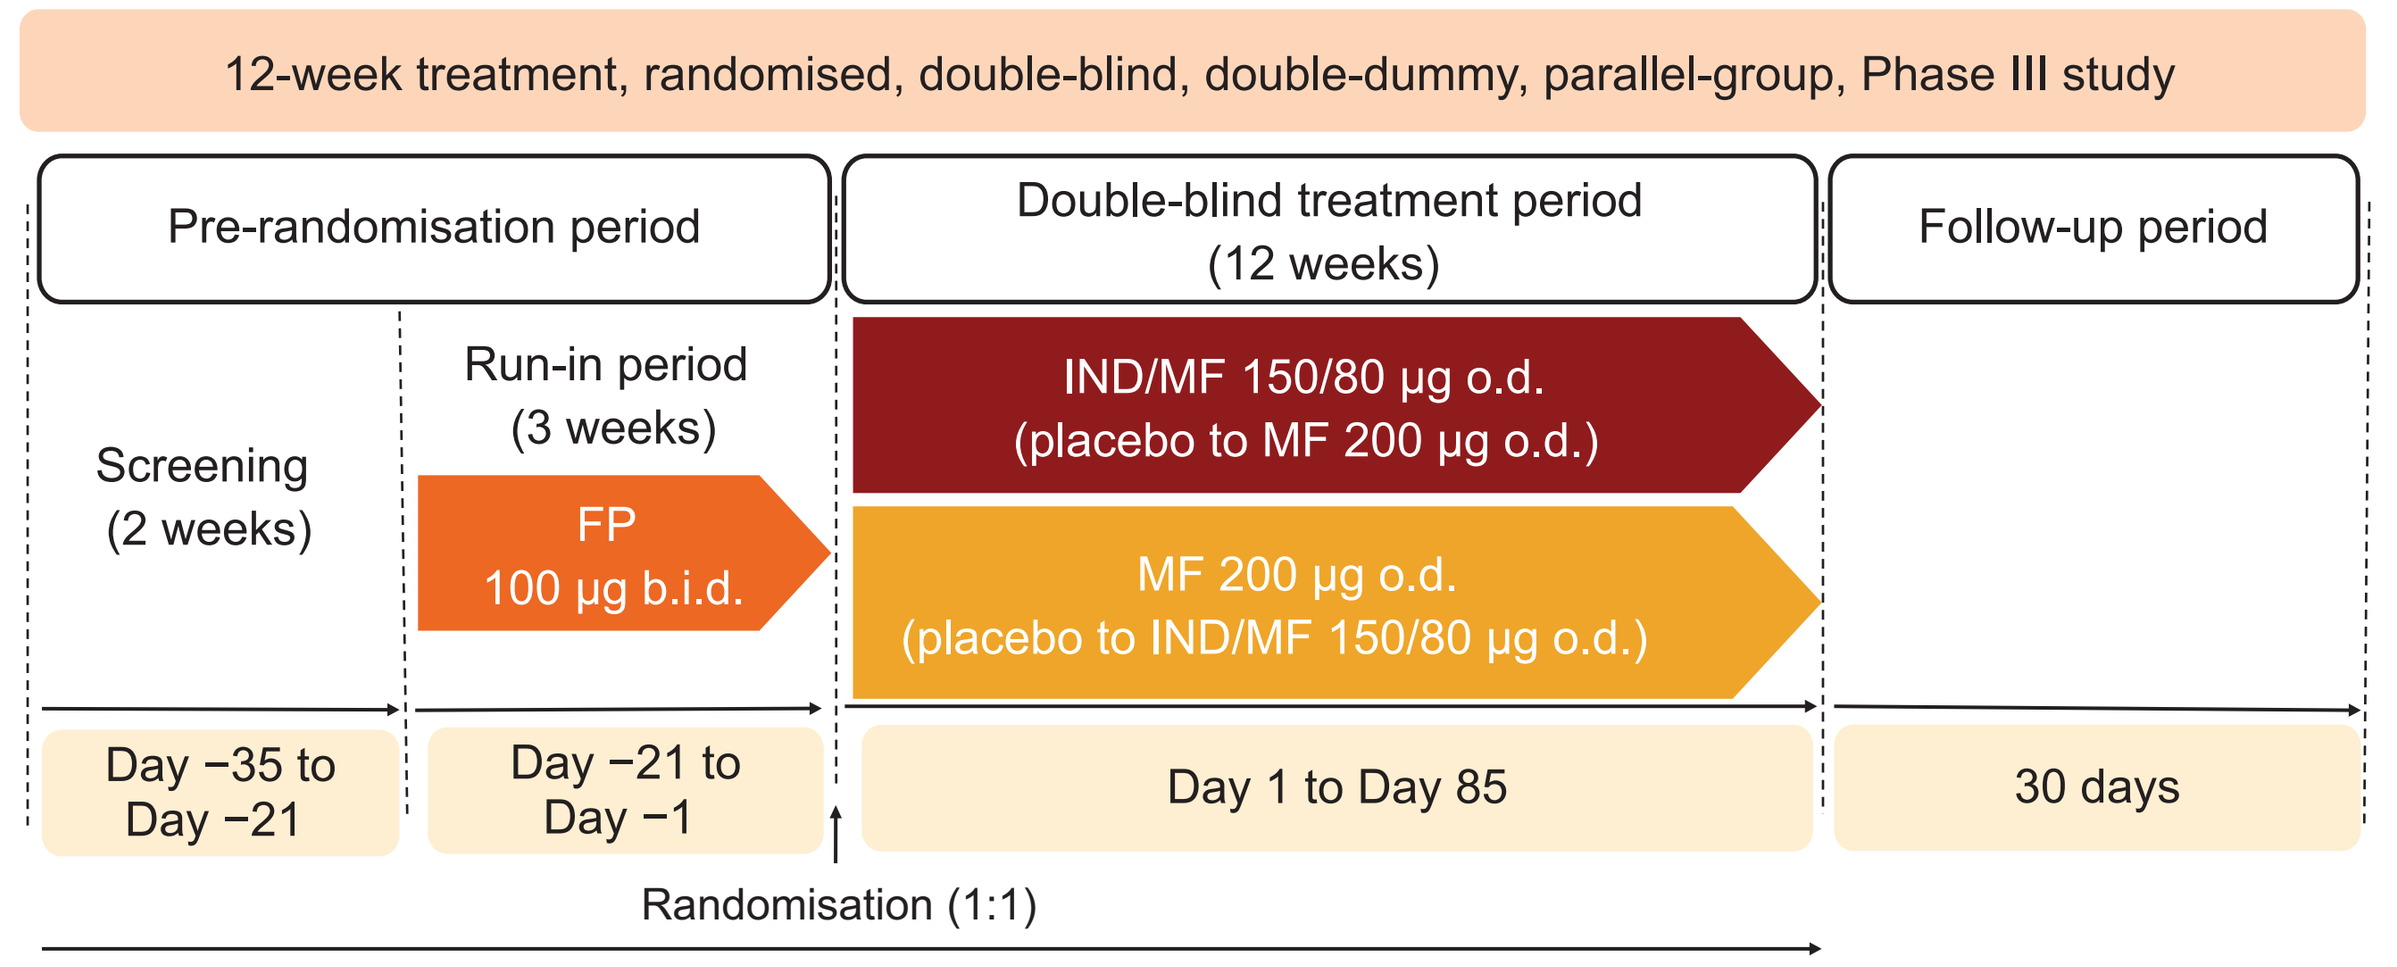
12-week treatment, randomised, double-blind, double-dummy, parallel-group, Phase III study
Pre-randomisation period
Double-blind treatment period (12 weeks)
Follow-up period
Screening (2 weeks)
Run-in period (3 weeks)
FP 100 µg b.i.d.
IND/MF 150/80 µg o.d. (placebo to MF 200 µg o.d.)
MF 200 µg o.d. (placebo to IND/MF 150/80 µg o.d.)
Day −35 to Day −21
Day −21 to Day −1
Day 1 to Day 85
30 days
Randomisation (1:1)
Rescue medication available

IND/MF was administered via Breezhaler® and MF via Twisthaler®
MF 200 µg o.d. via Twisthaler® is equivalent to MF 80 µg o.d. via Breezhaler®
b.i.d., twice daily; FP, fluticasone propionate; IND/MF, indacaterol acetate/mometasone furoate; MF, mometasone furoate; o.d., once-daily

Endpoints for evaluation of the efficacy of IND/MF (150/80 µg o.d.; [low-dose ICS]) versus MF 200 µg o.d. over 12 weeks of treatment

Primary endpoint

- Superiority of IND/MF (150/80 µg o.d.) to MF 200 µg o.d. in terms of trough FEV_1 after 12 weeks of treatment

Key secondary endpoint

- Superiority of IND/MF (150/80 µg o.d.) over MF 200 µg o.d. in terms of ACQ-7 score treatment difference after 12 weeks of treatment

Other secondary endpoints

- Proportion of patients with improvement in ACQ-7 score of at least 0.5 units (responder analysis)
- Morning and evening peak expiratory flow (PEF)
- Mean total daily symptom score, rescue medication use and nighttime awakenings (recorded via electronic diary)
- Asthma-related quality of life (asthma quality of life questionnaire [AQLQ] total score)
- Rate of moderate-to-severe exacerbations

Safety

- Safety assessments included adverse events, vital signs, ECG, laboratory analysis, serious asthma outcomes, other serious or clinically significant adverse events and deaths

Statistical analysis

- All efficacy analyses were performed using the full analysis set (FAS) population
 - FAS included all randomised patients who received at least one dose of study treatment
- All safety analyses were performed using the safety set
 - Safety set consisted of all patients who received at least one dose of study treatment
- Trough FEV_1 and ACQ-7 scores were analysed using mixed model for repeated measure (MMRM) model
- ACQ-7 responder analysis was analysed using a logistic regression model
- Percentage of nights with no nighttime awakenings was analysed using linear mixed model
- Rate of moderate-to-severe exacerbations was analysed using a generalised linear model assuming the negative binomial distribution
- All other efficacy assessments were evaluated using analysis of covariance (ANCOVA) model

Results

Table 1. Baseline demographics and clinical characteristics (randomised set)

Characteristic	IND/MF 150/80 µg o.d. (N = 398)	MF 200 µg o.d. (N = 404)	Total (N = 802)
Age, years	46.1 ± 16.26	45.1 ± 16.27	45.6 ± 16.26
Adolescents, n (%)	31 (7.8)	33 (8.2)	64 (8.0)
Adults, n (%)	367 (92.2)	371 (91.8)	738 (92.0)
Men, n (%)	151 (37.9)	163 (40.3)	314 (39.2)
Duration of asthma, years	14.4 ± 13.18	13.6 ± 12.48	14.0 ± 12.83
Baseline ACQ-7 score	2.24 ± 0.40	2.30 ± 0.39	2.27 ± 0.39
Prior asthma treatment, n (%)			
Low-dose ICS	177 (44.5)	167 (41.3)	344 (42.9)
Medium- to high-dose ICS	1 (0.3)	0	1 (0.1)
Low-dose ICS/LABA	217 (54.5)	232 (57.4)	449 (56.0)
Medium- to high-dose ICS/LABA	3 (0.8)	0	3 (0.4)
Missing	0	5 (1.2)	5 (0.6)
FEV_1 pre-bronchodilator (L)	2.24 ± 0.64	2.22 ± 0.58	2.23 ± 0.61
FEV_1 pre-bronchodilator (% predicted)	73.3 ± 7.57	72.2 ± 7.63	72.7± 7.61
FEV_1 reversibility (% increase)	20.6 ± 11.66	20.7 ± 11.94	20.7 ± 11.79

Data presented as mean ± standard deviation, unless otherwise specified.
ACQ-7, asthma control questionnaire; FEV_1, forced expiratory volume in one second; ICS, inhaled corticosteroid; IND/MF, indacaterol acetate/mometasone furoate; LABA, long-acting β_2-agonist; MF, mometasone furoate; o.d., once-daily

Primary endpoint: Trough FEV_1 at 12 weeks

Figure 2. IND/MF showed statistically significant improvement in trough FEV_1 compared with MF after 12 weeks of treatment

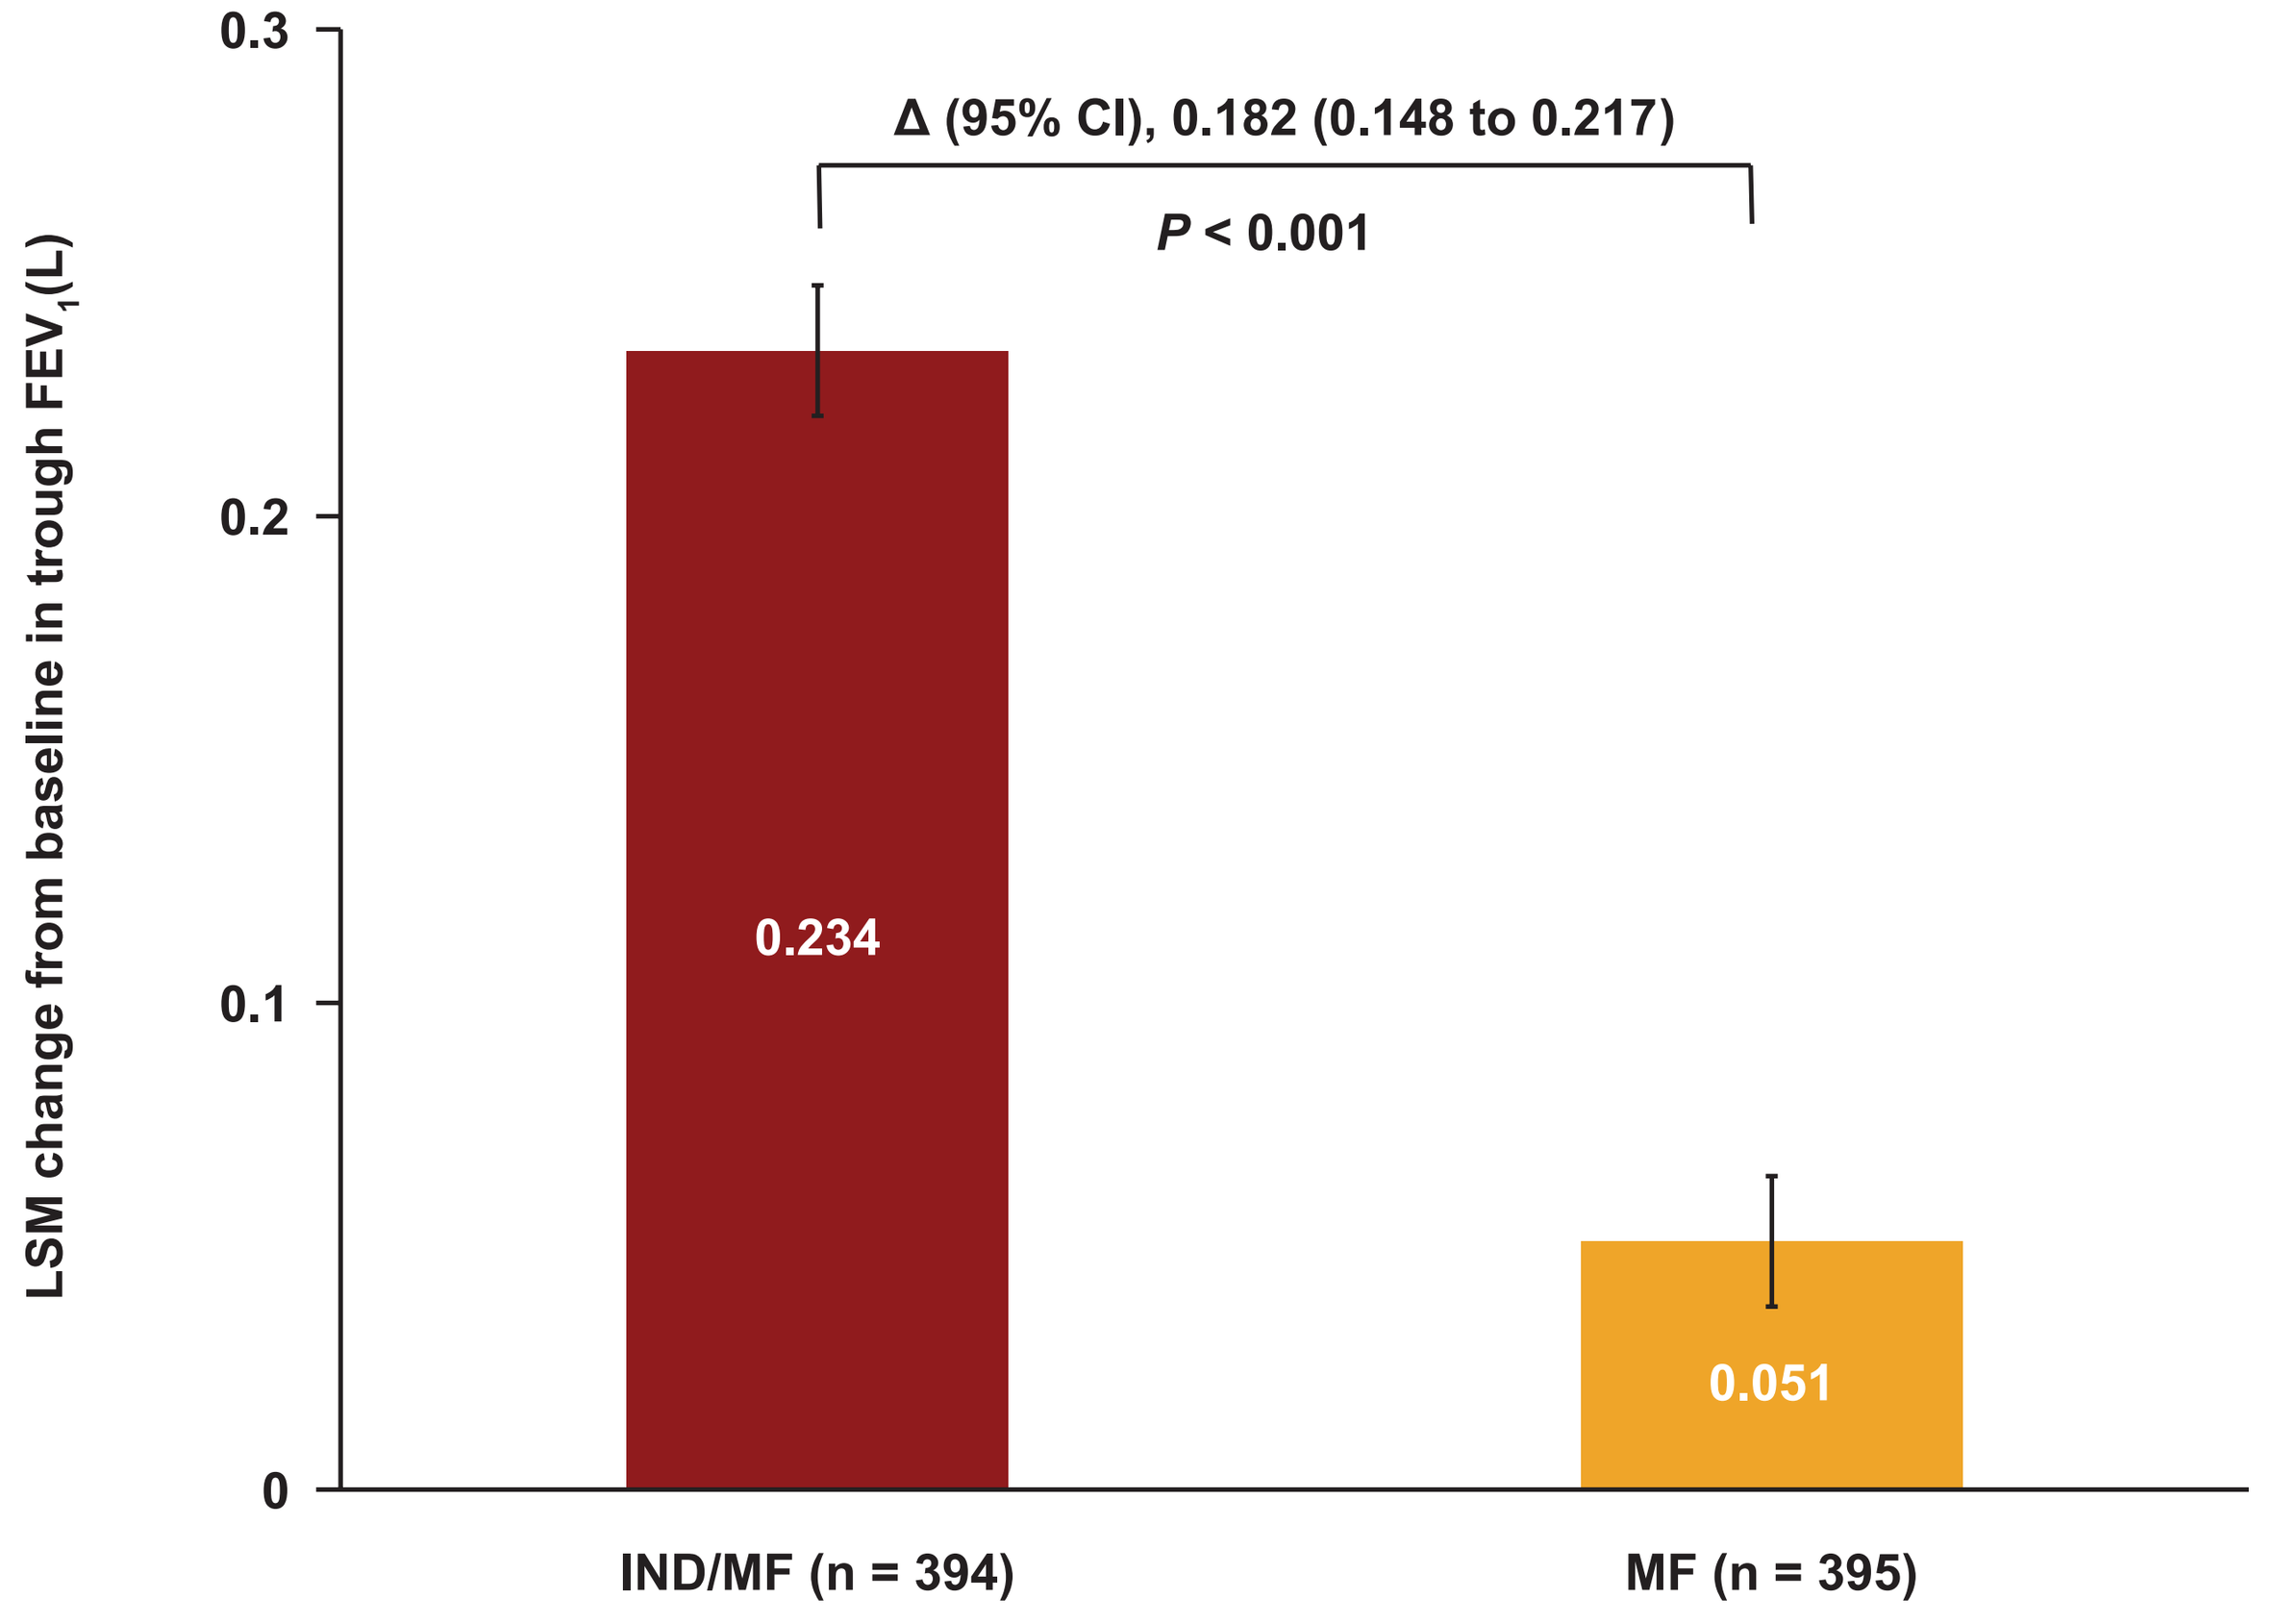
Δ (95% CI), 0.182 (0.148 to 0.217)
P < 0.001
LSM change from baseline in trough FEV1 (L)
0.3
0.2
0.1
0
0.234
0.051
IND/MF (n = 394)
MF (n = 395)

Data are presented as LSM ± SE. Error bars represent SE values
Δ, LSM treatment difference; CI, confidence interval; FEV_1, forced expiratory volume in 1 second; IND/MF, indacaterol/ mometasone furoate 150/80 µg o.d.; LSM, least squares mean; MF, mometasone furoate 200 µg o.d.; o.d., once-daily; SE, standard error

Figure 3. Trough and pre-dose trough FEV_1 was consistently higher with IND/MF at all time-points compared to MF over 12 weeks

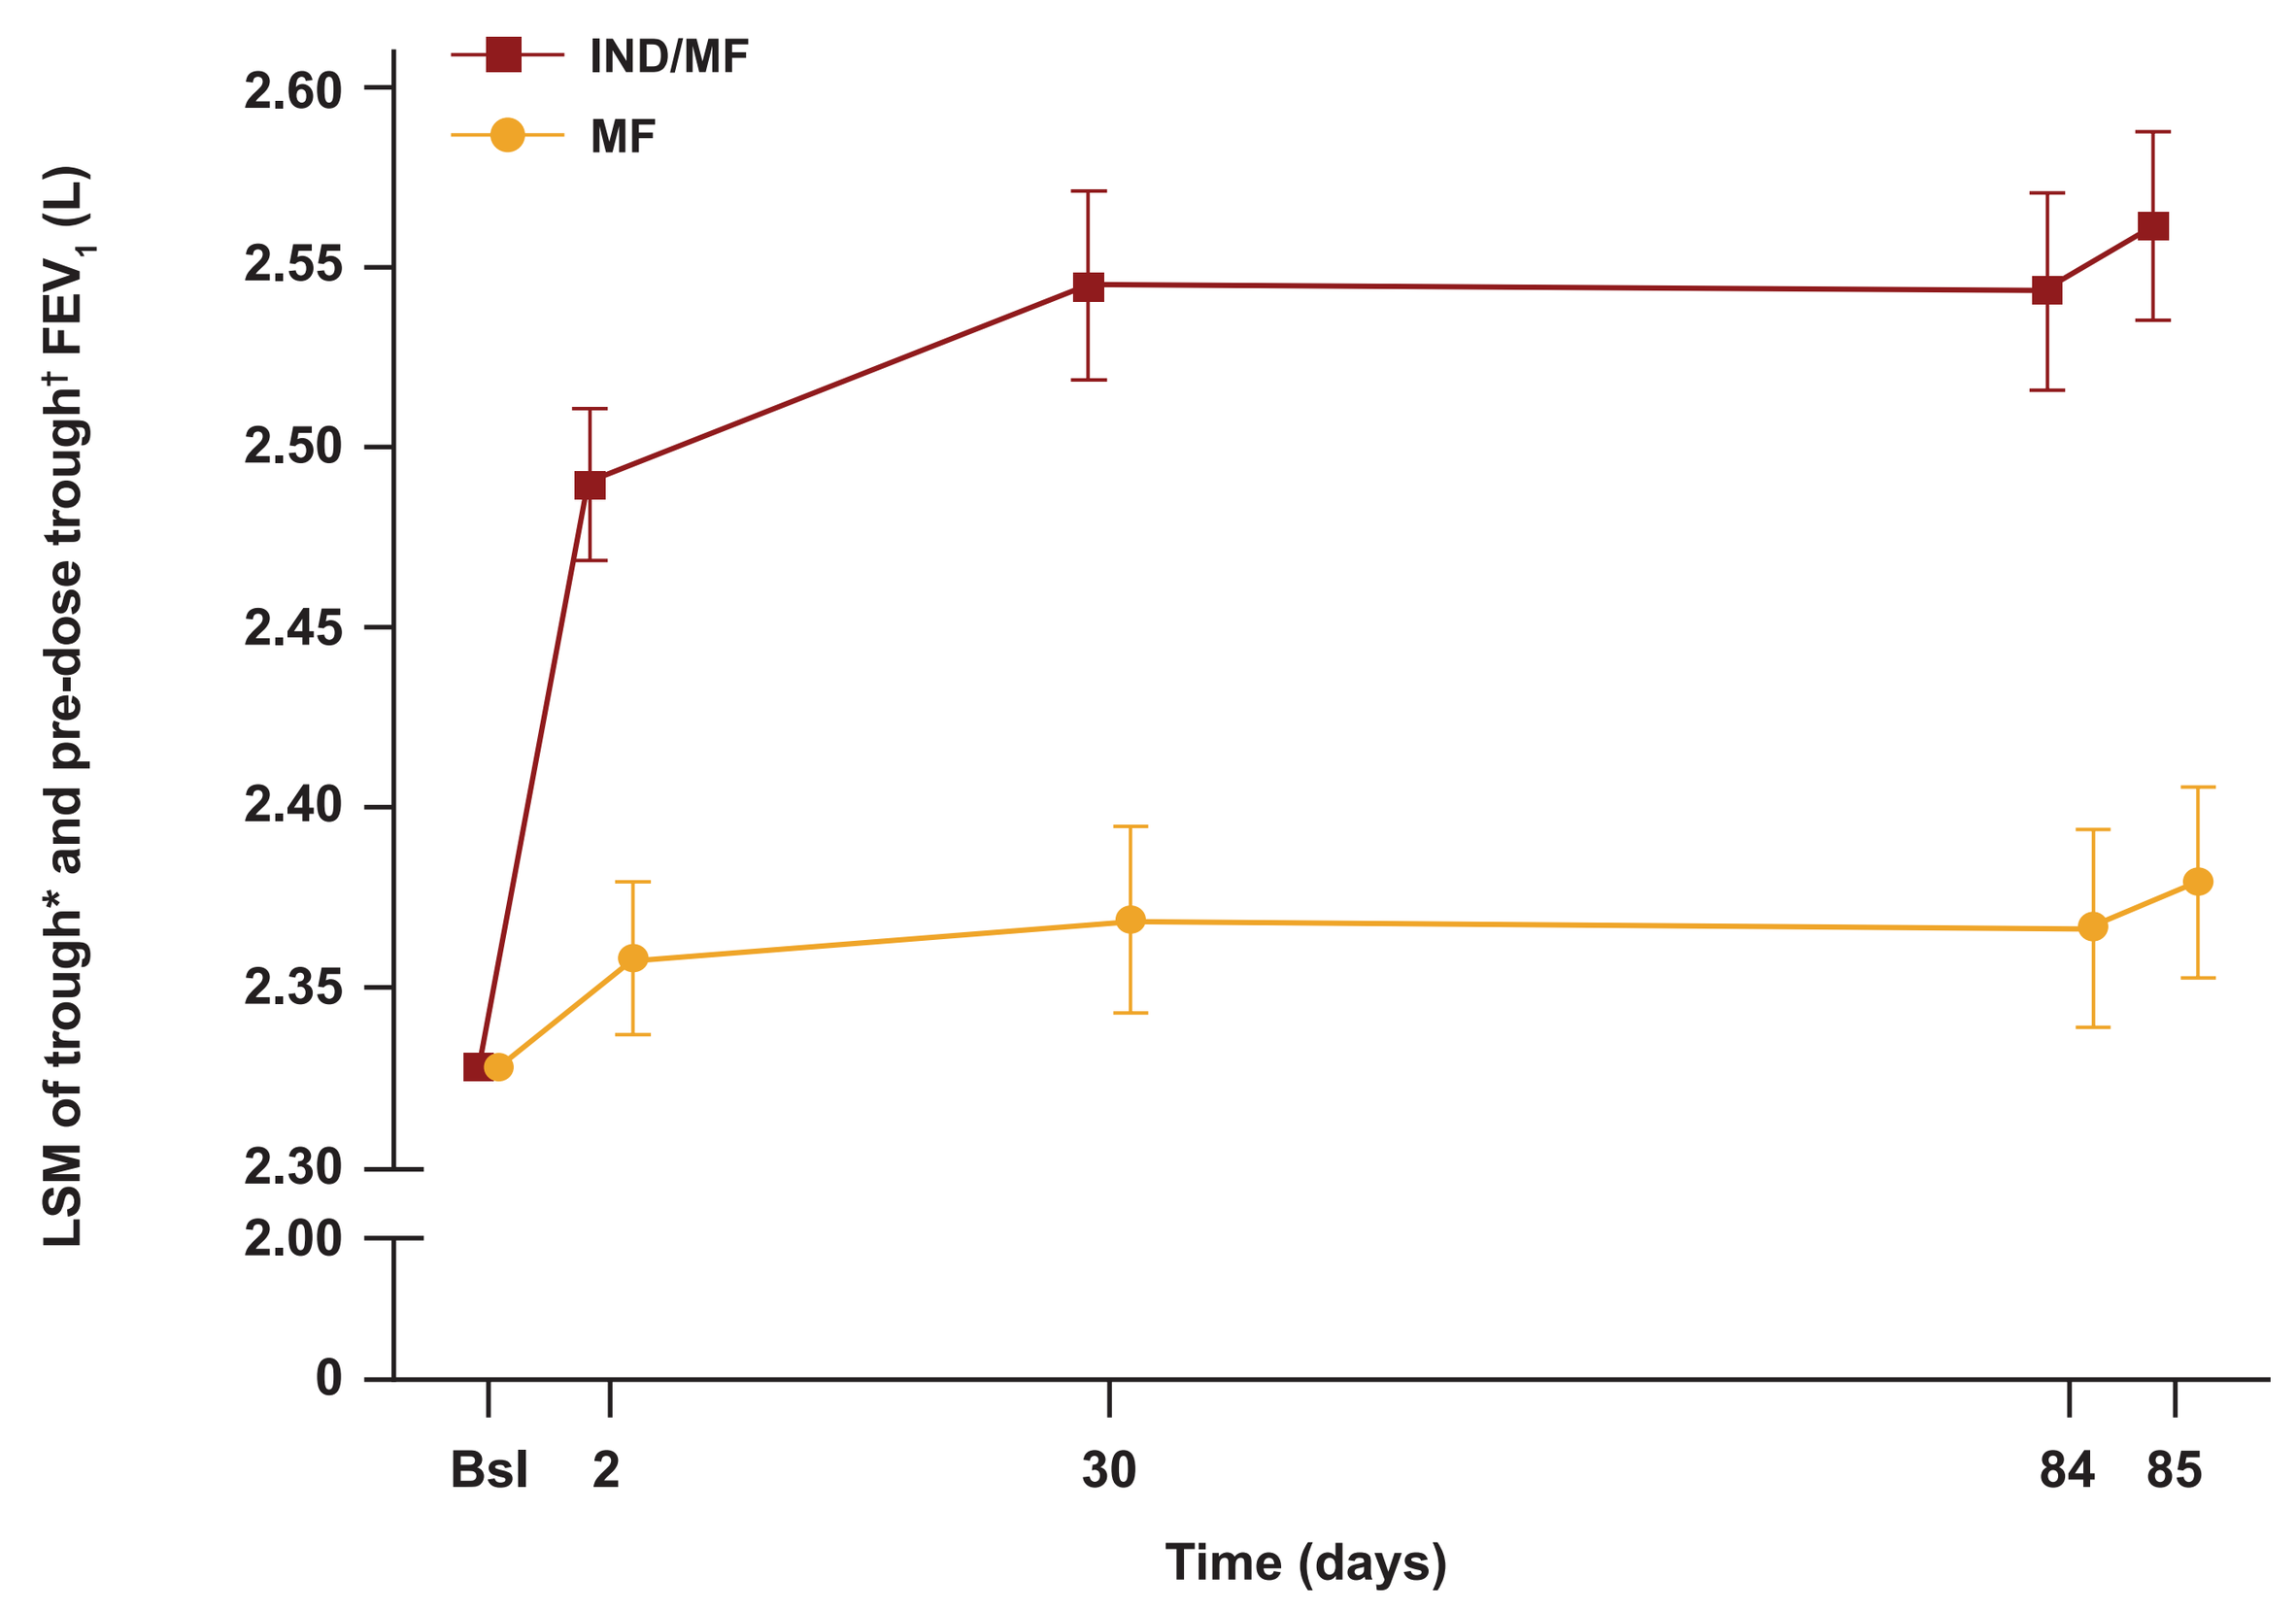
IND/MF
MF
LSM of trough* and pre-dose trough† FEV1 (L)
2.60
2.55
2.50
2.45
2.40
2.35
2.30
2.00
0
Bsl
2
30
84
85
Time (days)

*Trough FEV_1 is defined as average of the two FEV_1 measurements at 23 hr 15 min and 23 hr 45 min post-evening dose on Day 2 and Day 85
†Pre-dose trough FEV_1 is defined as average of the two FEV_1 measurements at 45 min and 15 min pre-evening dose on Day 30 and Day 84
Data are presented as LSM ± 95% CI. Error bars represent 95% CI
Bsl, baseline; CI, confidence interval; FEV_1, forced expiratory volume in 1 second; IND/MF, indacaterol/ mometasone furoate 150/80 µg o.d.; LSM, least squares mean; MF, mometasone furoate 200 µg o.d.; o.d., once-daily

Key secondary endpoint: ACQ-7 at Week 12

Figure 4. At Week 12, IND/MF treatment showed statistically significant improvements in asthma control from baseline

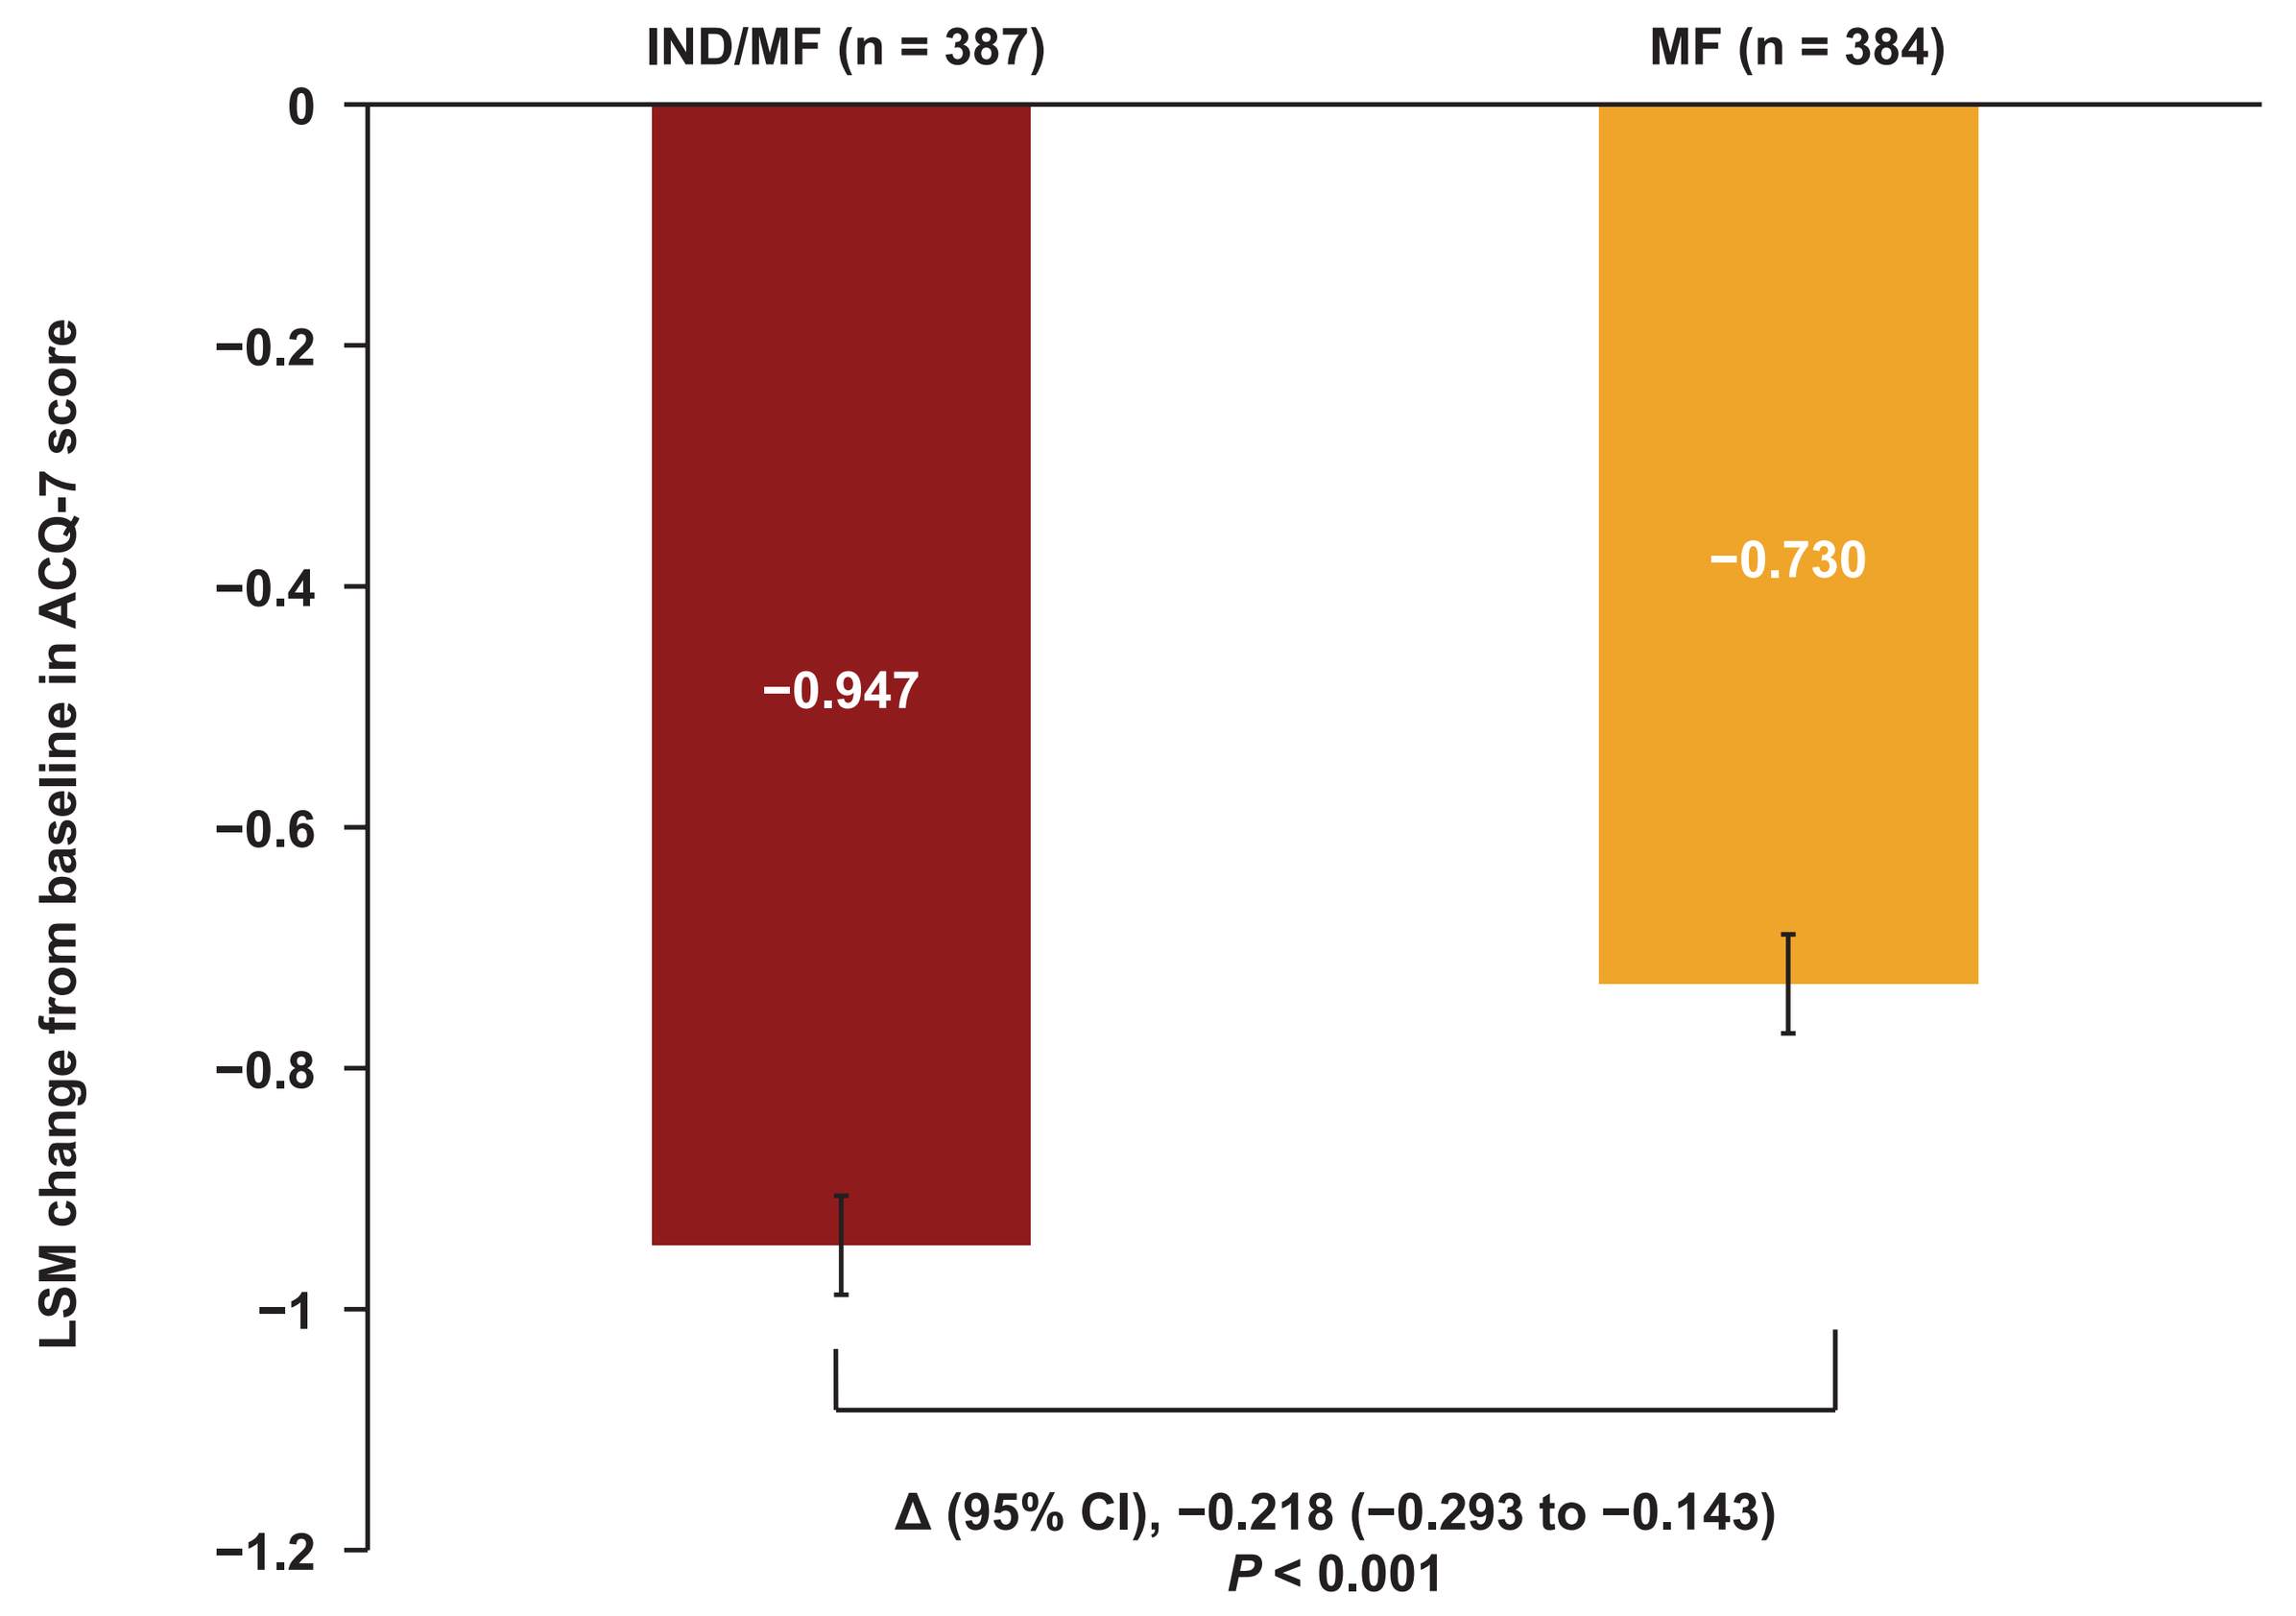
IND/MF (n = 387)
MF (n = 384)
LSM change from baseline in ACQ-7 score
0
−0.2
−0.4
−0.6
−0.8
−1
−1.2
−0.947
−0.730
Δ (95% CI), −0.218 (−0.293 to −0.143)
P < 0.001

Data are presented as LSM ± SE. Error bars represent SE values
ACQ-7, asthma control questionnaire; CI, confidence interval; IND/MF, indacaterol/ mometasone furoate 150/80 µg o.d.; LSM, least squares mean; MF, mometasone furoate 200 µg o.d.; o.d., once-daily; SE, standard error

ACQ-7 responder analysis

Figure 5. Higher proportion of patients achieved improvement of at least 0.5 units (MCID) in ACQ-7 score with IND/MF compared with MF after 12 weeks

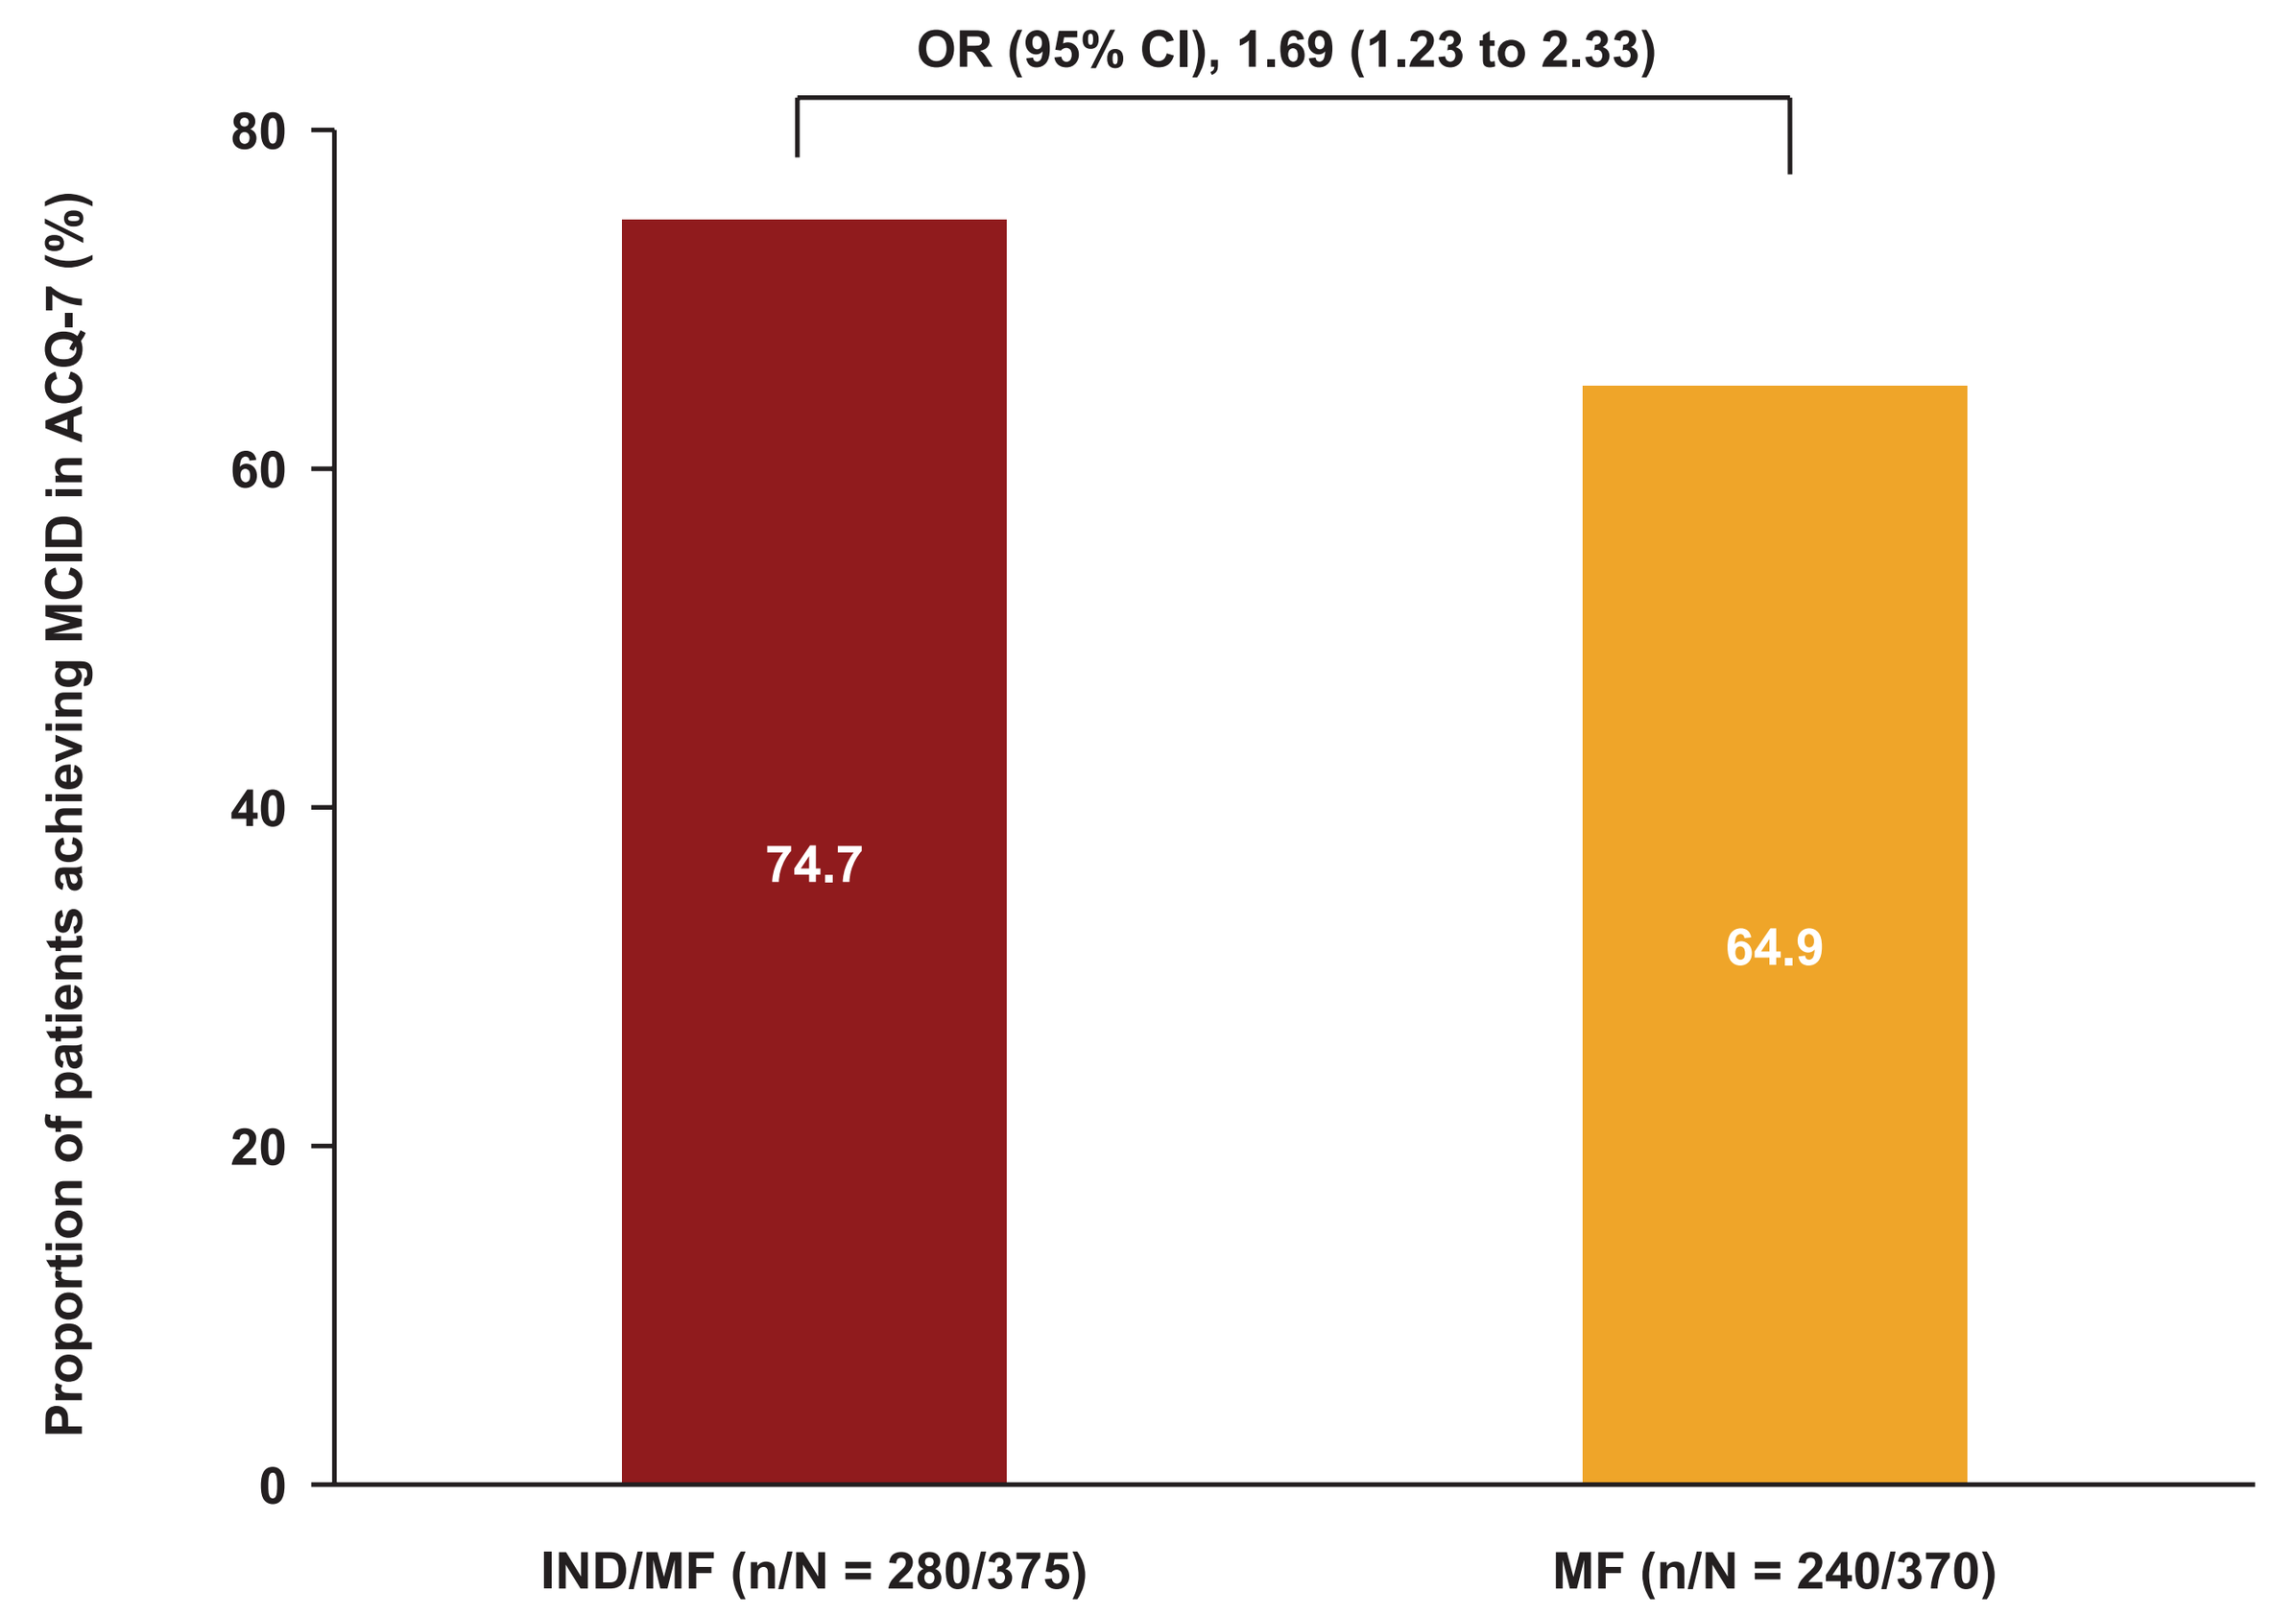
OR (95% CI), 1.69 (1.23 to 2.33)
Proportion of patients achieving MCID in ACQ-7 (%)
80
60
40
20
0
74.7
64.9
IND/MF (n/N = 280/375)
MF (n/N = 240/370)

n, The number of patients with an improvement of ≥0.5 units, i.e. a decrease of ≥0.5 units in ACQ-7 from baseline;
N, The number of patients with data at the respective visit.
Data are presented as LSM ± SE. Error bars represent SE values
ACQ-7, asthma control questionnaire; CI, confidence interval; IND/MF, indacaterol/ mometasone furoate 150/80 µg o.d.; MF, mometasone furoate 200 µg o.d.; o.d., once-daily; OR, odds ratio; SE, standard error; MCID, minimal clinically important difference

Table 2. Other secondary endpoints assessed over 12 weeks of treatment

Secondary endpoint	IND/MF 150/80 µg o.d. vs MF 200 µg o.d. Treatment difference (95% CI)
Mean morning PEF, L/min	27.2 (22.1 to 32.4)
Mean evening PEF, L/min	26.1 (21.0 to 31.2)
Rescue medication use	−0.26 (−0.37 to −0.14)
Quality of life (AQLQ total score)	0.149 (0.064 to 0.234)
Mean total daily symptom score	−0.15 (−0.26 to −0.05)
Percentage of nights with no nighttime awakenings	4.8 (1.8 to 7.7)
Rate of moderate-to-severe exacerbations	RR: 0.25 (0.12 to 0.52)

AQLQ, asthma quality of life questionnaire; CI, confidence interval; IND/MF, indacaterol/ mometasone furoate 150/80 µg o.d.; LSM, least squares mean; MF, mometasone furoate 200 µg o.d.; o.d., once-daily; PEF, peak expiratory volume; RR, rate ratio

Safety

- The overall incidence of adverse events (AEs) was lower in patients treated with IND/MF 150/80 µg o.d. (32.3%) compared with the MF (38.3%)
- One serious asthma outcome (asthma-related hospitalisation) was reported in the MF group and none in IND/MF group
- Overall, the incidence of serious AEs was low and comparable between the treatment groups (1.3% in the IND/MF group and 1.8% in the MF group)
- No death was reported in any treatment group throughout the study

Conclusions

- To our knowledge, this is the first study to compare a low-dose ICS/LABA FDC o.d. to low-dose ICS monotherapy in adult and adolescent patients with inadequately controlled asthma
- IND/MF 150/80 µg o.d. significantly improved trough FEV_1 and asthma control versus MF 200 µg o.d. in adult and adolescent patients with inadequately controlled asthma
- This study provided evidence for benefits across a range of lung function and symptomatic endpoints including: morning and evening PEF, AQLQ total score, mean daily total symptom score, rescue medication use, nighttime awakenings and rate of moderate-to-severe exacerbations
- Safety profile was comparable between the two treatment groups
- Overall, results support the use of IND/MF 150/80 µg o.d. as a potential treatment in adult and adolescent patients with asthma, inadequately controlled on low-dose ICS (+/- controller)

References

1. Global strategy for asthma management and prevention, Global Initiative for Asthma (GINA) 2019. Available from: http://www.ginasthma.org/

Acknowledgements

The authors were assisted in the preparation of the poster by Hasitha Shilpa Anantaraju and Jisha John (Novartis)

Copyright © 2019 Novartis Pharma AG. All rights reserved

The study was funded by Novartis Pharmaceuticals Corporation, East Hanover, New Jersey, USA

Poster presented at the European Respiratory Society International Congress, September 28–October 2, 2019, Madrid, Spain

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors

Scan this QR code