Inhibition of $A_{2A}R$ by AZD4635 induces anti-tumor immunity alone and in combination with anti-PD-L1 in preclinical models

Alexandra Borodovsky[1], Yanjun Wang[1], Minwei Ye[1], Nanhua Deng[1], Joseph C Shaw[1], Kris Sachsenmeier[1], Kelly Goodwin[1], James D Clarke[1], Richard Goodwin[1], Nicole Strittmatter[1], Carl Hay[2], Vasu Sah[1], Deborah Lawson[1], Corinne Reimer[1], Miles Congreve[3], Jon S Mason[3], Fiona H Marshall[3], Paul Lyne[1], Rich Woessner[1]

[1]AZ IMED Biotech Unit, Boston, MA USA , [2] MedImmune, Gaithersburg, MD USA, [3]Heptares Therapeutics Ltd, Welwyn Garden City, Hertfordshire, UK

3751

Abstract

Adenosine signaling through the high affinity adenosine 2A receptor ($A_{2A}R$) on immune cells elicits a range of immunosuppressive effects which can promote tumor growth and limit the effectiveness of immune-mediated anti-tumor responses. AZD4635 (HTL-1071) is a potent and selective oral $A_{2A}R$ antagonist, currently in a Phase 1 clinical trial as a single agent and in combination with durvalumab (anti-PD-L1 Ab) in patients with solid malignancies. In T cell IFNγ secretion cell assays, the EC_{50} of AZD4635 is dependent on adenosine concentrations however, full understanding of the impact of adenosine on AZD4635 mediated anti-tumor responses requires characterization of intratumoral adenosine concentration and spatial heterogeneity. Using a novel LC/MS based method, measurement of intratumoral adenosine concentrations in a panel of syngeneic tumor models demonstrated a wide range of adenosine levels. Additionally, measurement of intratumoral adenosine by DESI-MS demonstrated that adenosine levels are spatially heterogeneous. The therapeutic benefit of $A_{2A}R$ blockade alone and in combination with anti-PD-L1 was evaluated in syngeneic mouse tumor models with varying adenosine concentrations. Treatment with AZD4635 alone and in combination with an anti-PD-L1 Ab led to a reduction in tumor growth in both adenosine high and adenosine low syngeneic tumor models. Tumors harvested from the treated mice exhibited increases in intratumoral T cell infiltration and increases in expression of co-stimulatory molecules on APCs. These results demonstrate that AZD4635 is a potent and selective $A_{2A}R$ inhibitor, and that blockade of $A_{2A}R$ signaling with an inhibitor such as AZD4635 can reduce tumor burden and enhance antitumor immunity as a single agent and in combination with an anti-PD-L1 antibody in preclinical tumor models.

Introduction

- Accumulation of extracellular adenosine within the microenvironment is a strategy exploited by tumors to escape immune surveillance.
- Adenosine signaling through the high affinity adenosine 2A receptor ($A_{2A}R$) on immune cells elicits a range of immunosuppressive effects
- Blockade of the A_{2A} receptor can reverse adenosine mediated immune suppression to enhance anti-tumor immunity

Results

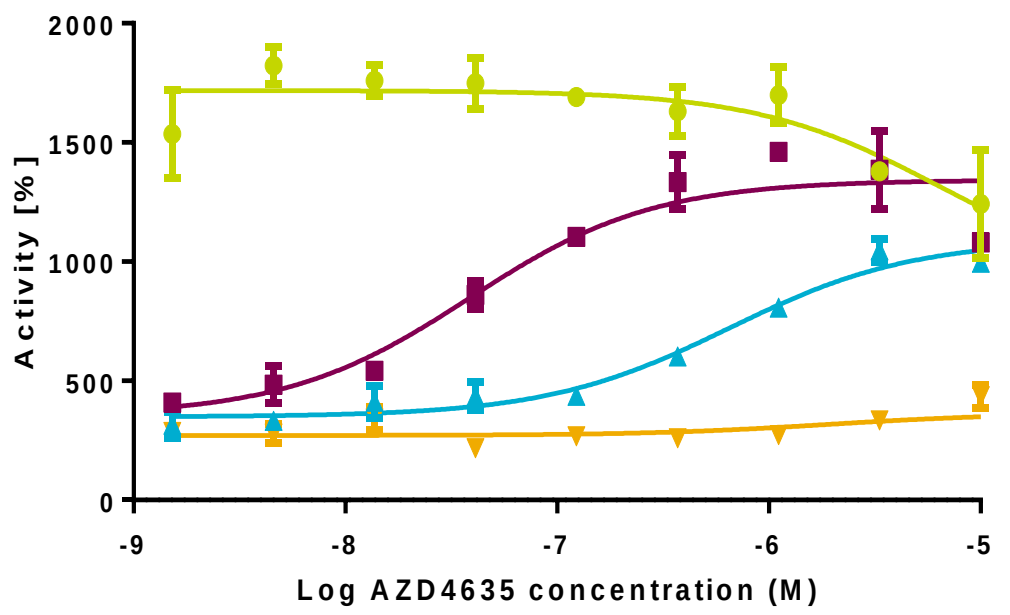
Activity [%]
Log AZD4635 concentration (M)

Key	NECA (µM)	AZD4635 EC_{50} (µM) ±SD
	10	>10
	1	0.598 ± 0.067
	0.1	0.053 ± 0.015
	UT	N/A

AZD4635 reverses adenosine mediated T cell suppression. EC_{50} (µM) of AZD4635 in a mouse CD8a+ T cell IFNγ secretion assay. The EC_{50}s of AZD4635 for reversal of NECA-mediated suppression of IFNγ secretion were determined across a range of NECA concentrations. The activity was calculated as ((Sample Data – Blank) / (Untreated Data - Blank))*100. EC_{50} was calculated as the compound concentration that gave half-maximal response. Error bars represent SD.

Results

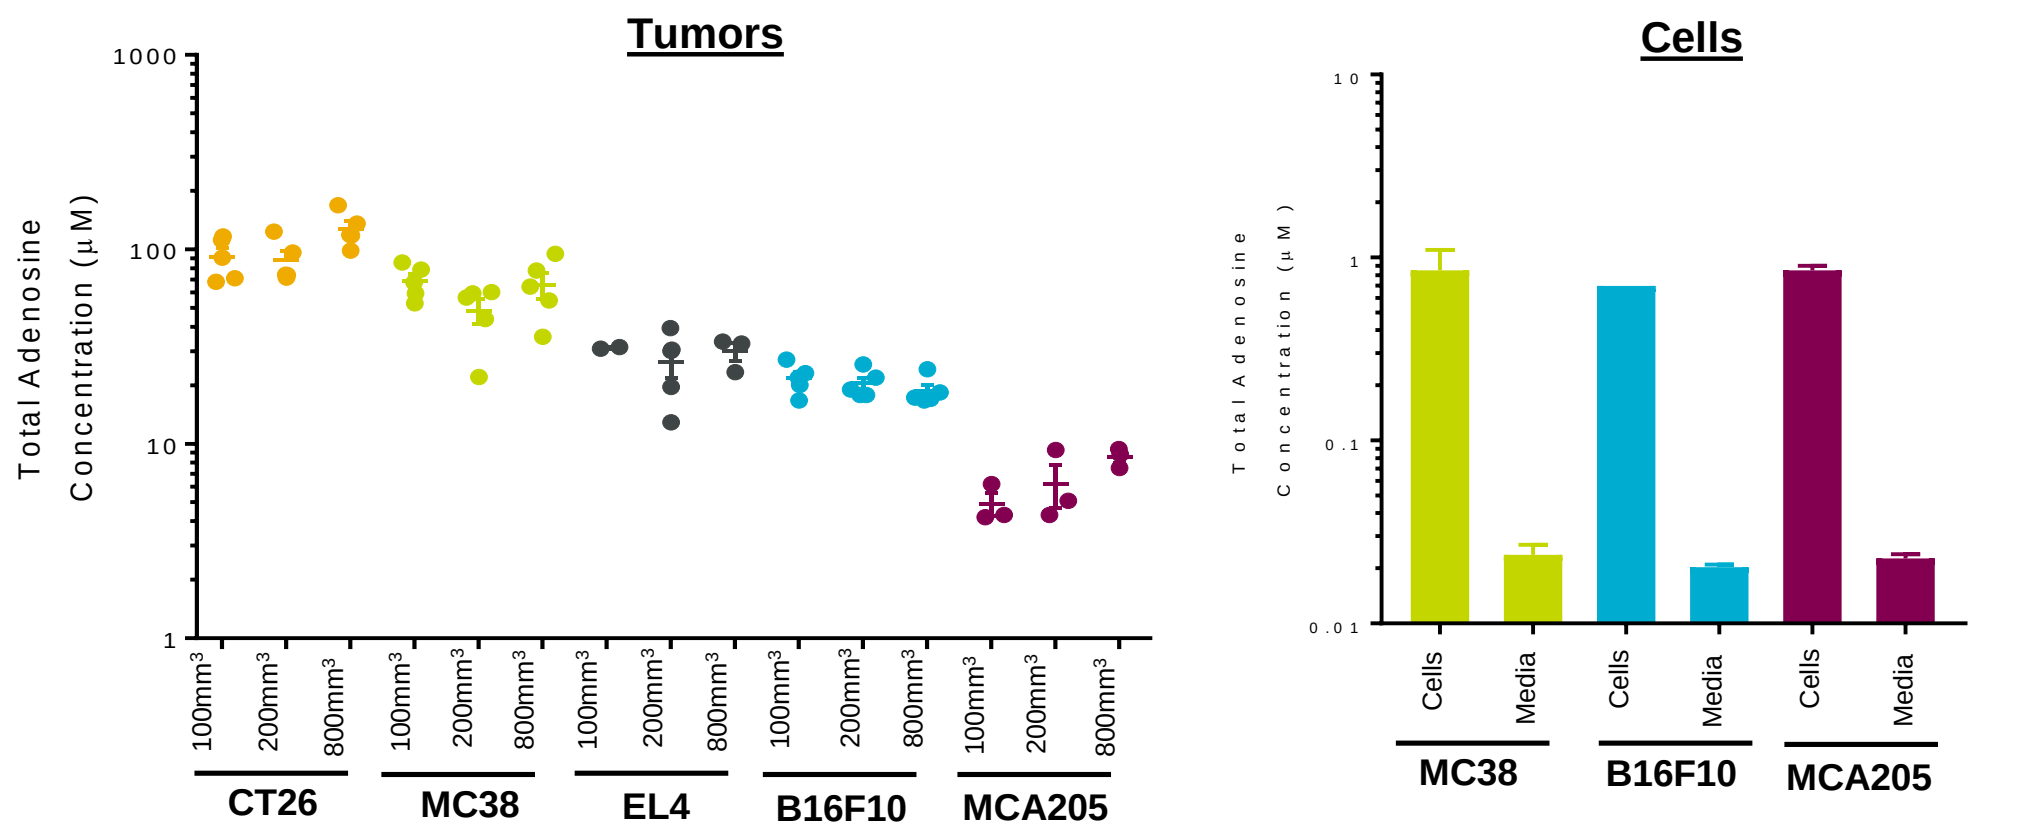
Tumors
Cells
Total Adenosine Concentration (µM)
CT26
MC38
EL4
B16F10
MCA205
Cells
Media

Total adenosine concentrations are high in mouse syngeneic tumors compared to corresponding cell lines. Average total adenosine concentration was measured by a novel LC/MS/MS assay in a panel of syngeneic tumors of varying volumes and in corresponding cell lines and conditioned media. Bars represent an average of 3 replicates, (error bars represent SEM).

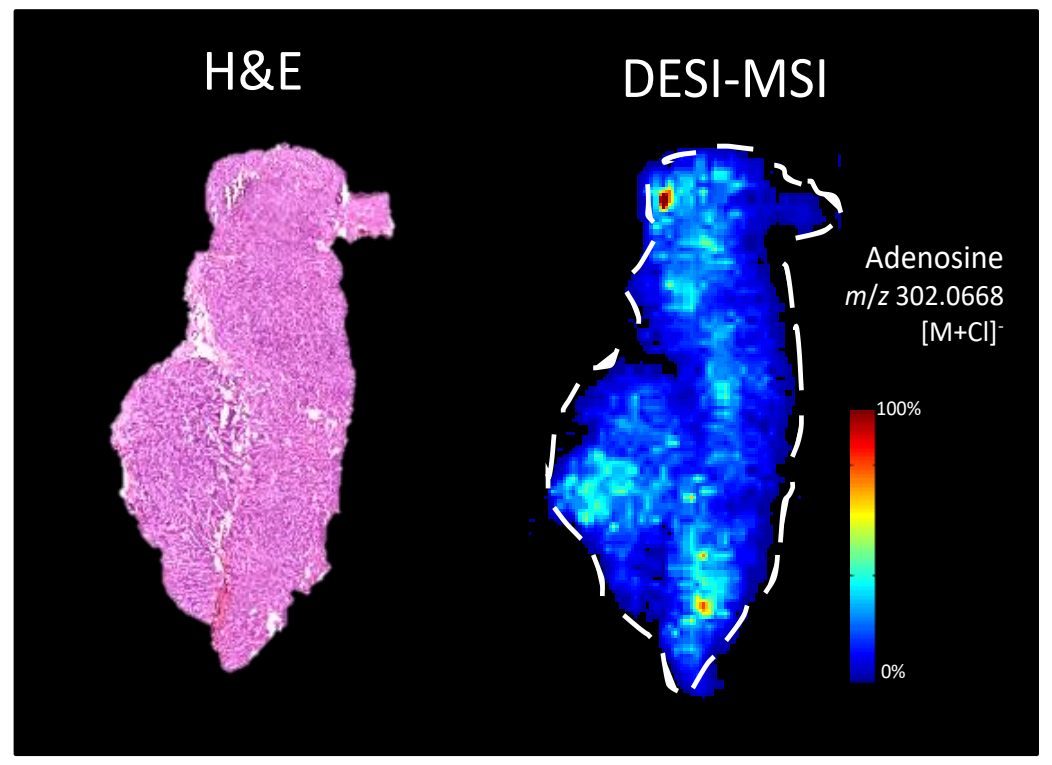
H&E
DESI-MSI
Adenosine
m/z 302.0668
[M+Cl]-
100%
0%

Adenosine levels are spatially heterogeneous in syngeneic tumors. Accumulation of intratumoral adenosine in subcutaneous murine CT26 colorectal carcinoma tumors, measured by desorption electrospray ionisation - mass spectroscopy (DESI-MS) (right panel), demonstrated that adenosine levels were spatially heterogeneous, with "pockets" of adenosine high and low regions. Alignment of pockets of adenosine and of immune cells is therefore required for complete understanding of AZD4635 target coverage. Scale bar: arbitrary abundance threshold at 50%. H&E staining (left panel).

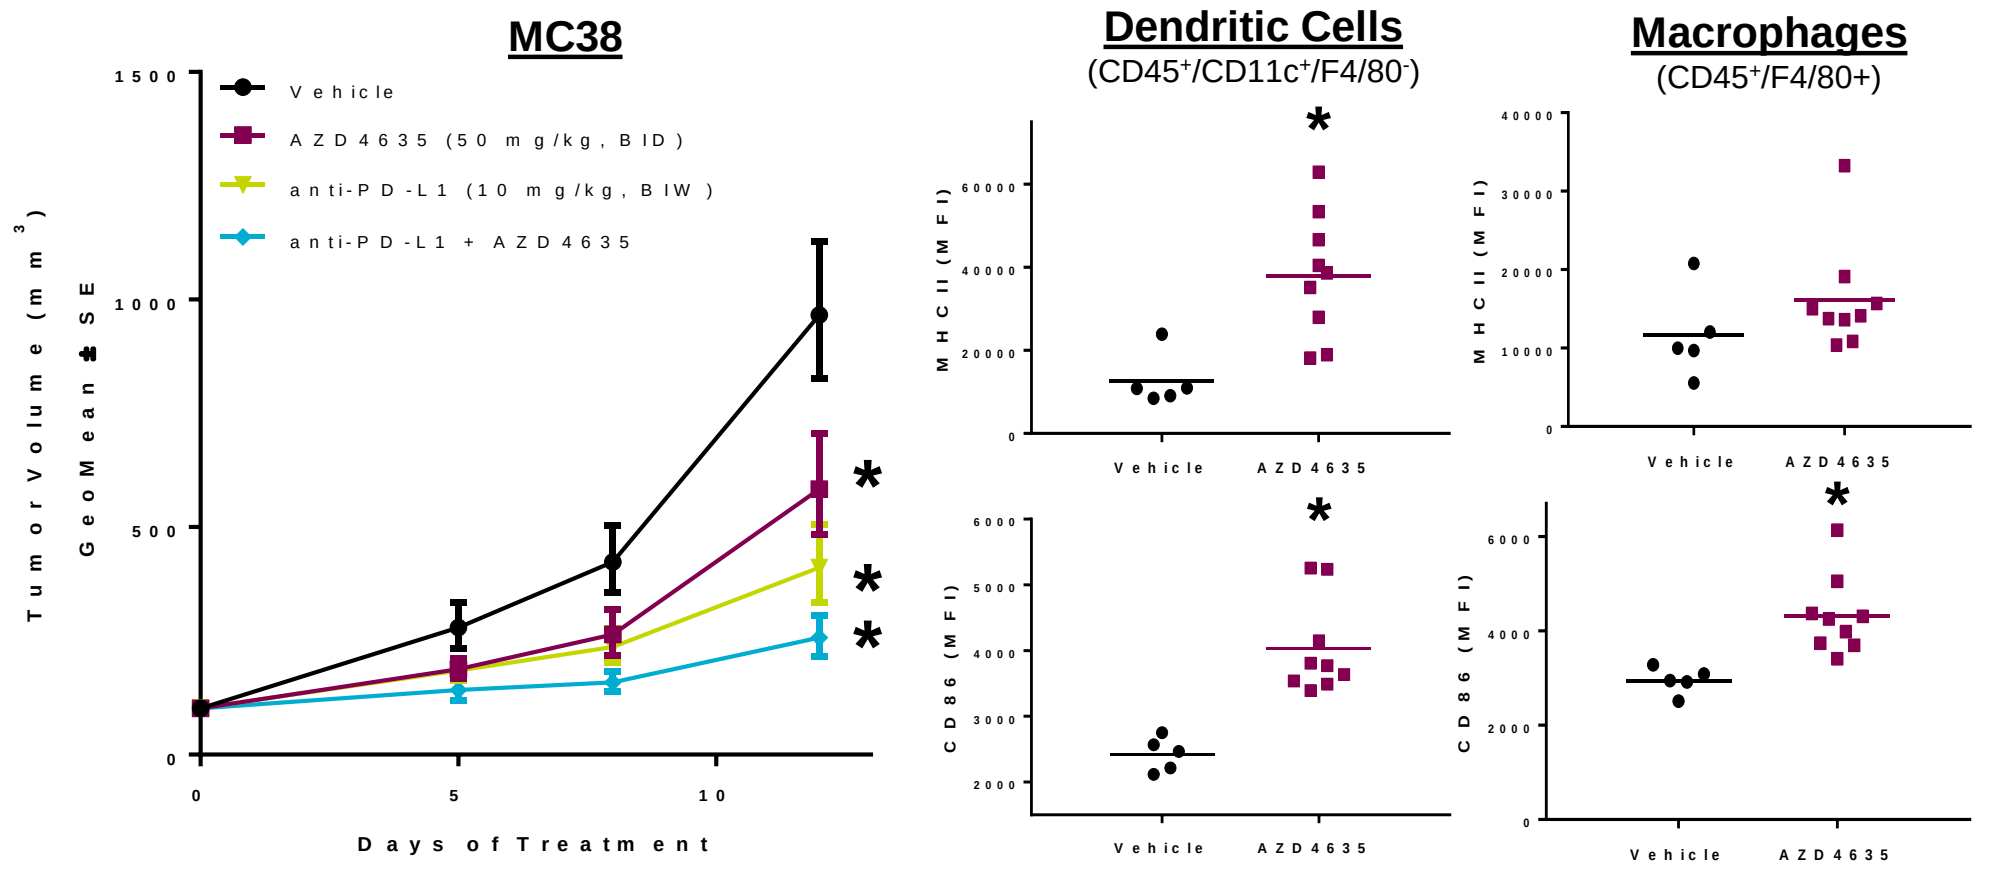
MC38
Vehicle
AZD4635 (50 mg/kg, BID)
anti-PD-L1 (10 mg/kg, BIW)
anti-PD-L1 + AZD4635
Tumor Volume (mm3) GeoMean ± SE
Days of Treatment
Dendritic Cells
(CD45+/CD11c+/F4/80-)
Macrophages
(CD45+/F4/80+)
MHCII (MFI)
CD86 (MFI)
Vehicle
AZD4635

AZD4635 exhibits antitumor activity an increases in expression of co-stimulatory molecules in an adenosine high syngeneic tumor model. The therapeutic benefit of $A_{2A}R$ blockade in an adenosine high model was evaluated in established MC38 syngeneic colorectal cancer in combination with anti-PD-L1 (IgG2). Inhibition of $A_{2A}R$ signaling by AZD4635 led to a reduction in tumor growth alone and in combination with anti-PD-L1 as well as increased expression of co-stimulatory markers (CD86) and markers of antigen presentation (MHCII) on dendritic cells and macrophages after 14 days of dosing (error bars represent SEM, *p<0.05)

Results

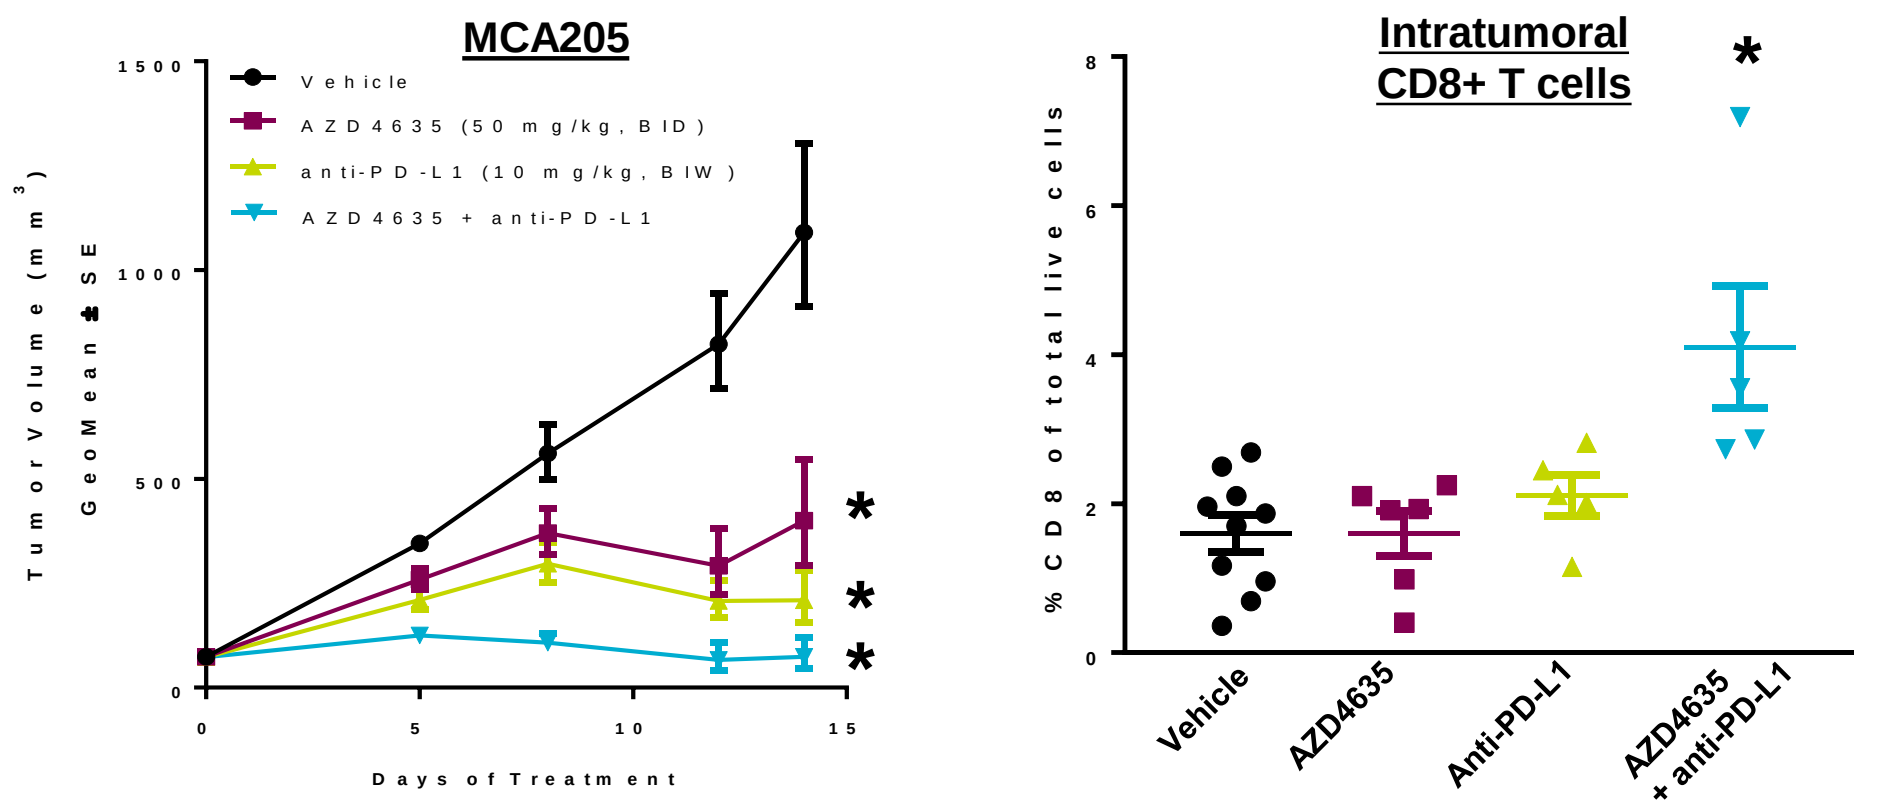
MCA205
Vehicle
AZD4635 (50 mg/kg, BID)
anti-PD-L1 (10 mg/kg, BIW)
AZD4635 + anti-PD-L1
Tumor Volume (mm3) GeoMean ± SE
Days of Treatment
Intratumoral CD8+ T cells
% CD8 of total live cells
Vehicle
AZD4635
Anti-PD-L1
AZD4635 + anti-PD-L1

AZD4635 exhibits antitumor activity and increases intratumoral lymphocyte infiltration in adenosine low syngeneic tumor models. The therapeutic benefit of $A_{2A}R$ blockade in an adenosine low model was evaluated in established MCA205 syngeneic fibrosarcoma cancer in combination with anti-PD-L1 (IgG1, D265A). AZD4635 led to a reduction of tumor growth alone and in combination with anti-PD-L1 as well as a significant increase of increased intratumoral CD8+ T cells following 14 days of dosing (Error bars represent SEM, *p<0.05).

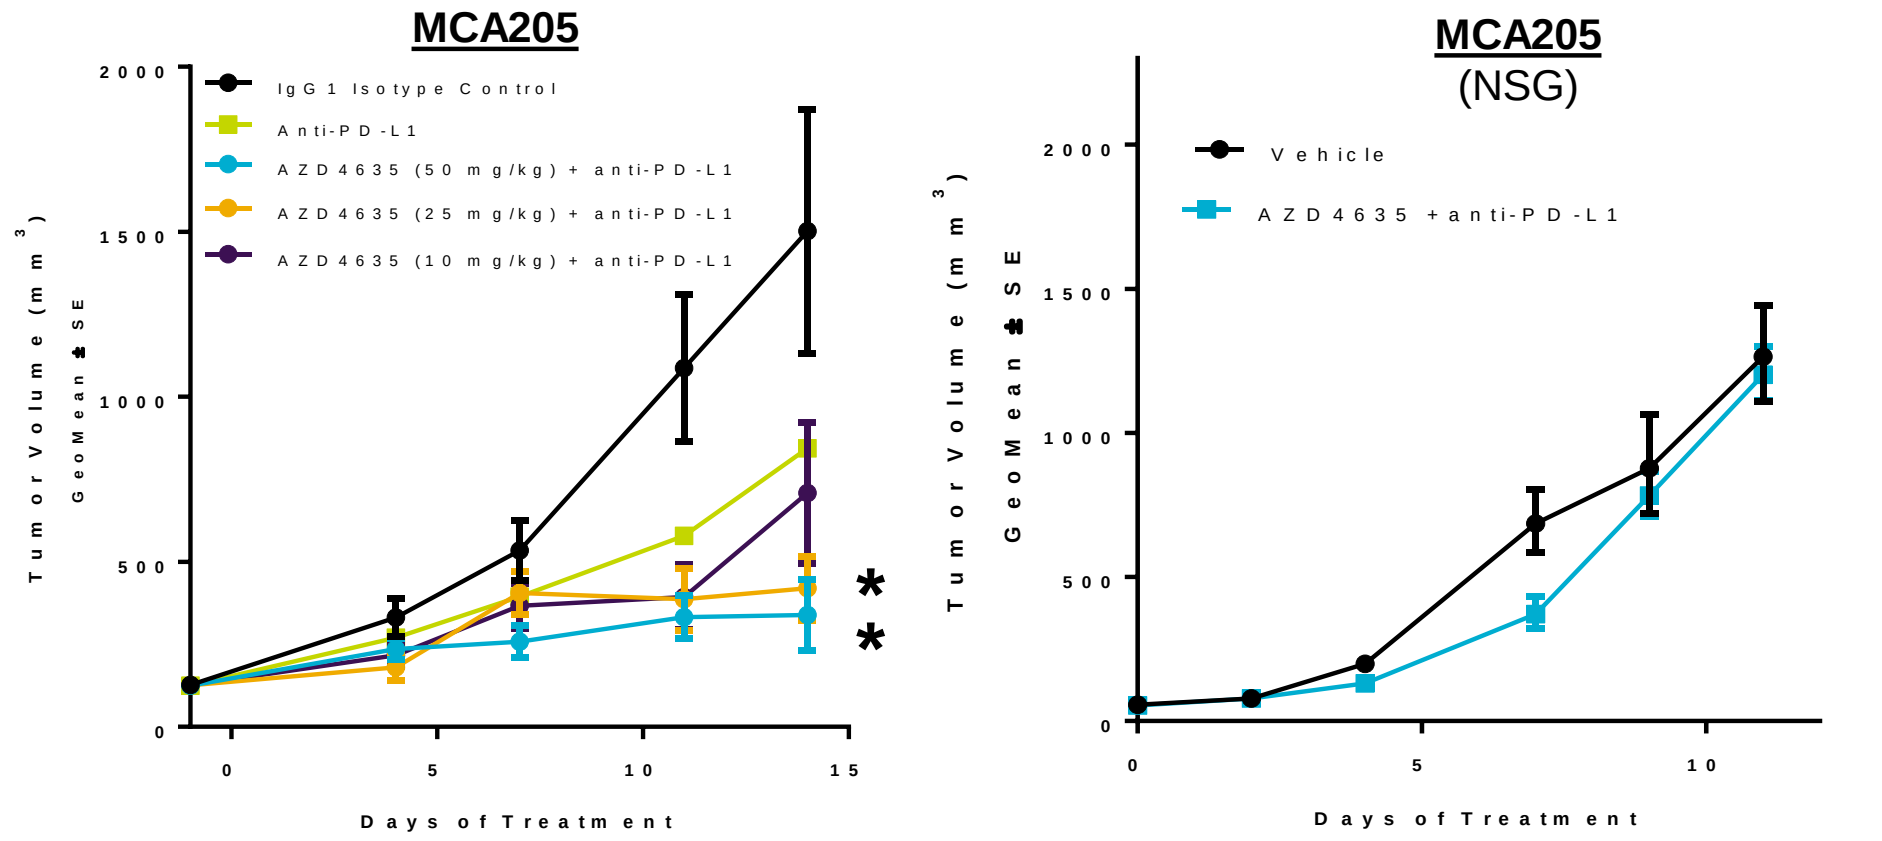
MCA205
IgG1 Isotype Control
Anti-PD-L1
AZD4635 (50 mg/kg) + anti-PD-L1
AZD4635 (25 mg/kg) + anti-PD-L1
AZD4635 (10 mg/kg) + anti-PD-L1
MCA205
(NSG)
Vehicle
AZD4635 + anti-PD-L1
Tumor Volume (mm3) GeoMean ± SE
Days of Treatment

AZD4635 exhibits dose dependent tumor growth inhibition, and inhibition is abrogated in immune deficient animals. AZD4635 led to a dose dependent reduction in tumor burden in combination with anti-PD-L1 (IgG1 D265A) in established MCA205 syngeneic fibrosarcoma tumors (left panel). To confirm the immune mediated mechanism, immune deficient NOD-*scid* IL2Rg^{null} (NSG) mice were implanted with subcutaneous MCA205 tumors and treated with AZD4635 and anti-PD-L1 (D265A). The anti-tumor activity was completely abrogated in NSG mice, confirming the immune mediated mechanism of tumor growth inhibition (right panel). Error bars represent SEM, *p<0.05.

Conclusions

- AZD4635 (HTL-1071) is an oral $A_{2A}R$ antagonist that reverses adenosine mediated T cell suppression
- Total adenosine levels are high and spatially heterogenous within syngeneic tumors. Spatial overlap of adenosine concentration and immune cell distribution is required to inform AZD4635 target coverage. Further exploration of target engagement by AZD4635 is ongoing.
- Treatment with AZD4635 alone and in combination with an anti-PD-L1 Ab led to a reduction in tumor growth in both adenosine high and adenosine low syngeneic tumor models
- Inhibition of $A_{2A}R$ signaling by AZD4635 led to an increase in expression of co-stimulatory markers on APCs and increased intratumoral infiltration of CD8+T cells in combination with anti-PD-L1
- AZD4635 is currently in a Phase 1 clinical trial as a single agent and in combination with durvalumab (anti-PD-L1 Ab) in patients with solid malignancies (NCT02740985)

Supported by AstraZeneca IMED Biotech Unit

Presented at the AACR Annual Meeting, Chicago IL., 17 April, 2018